14TH INTERNATIONAL CONGRESS OF INBORN ERRORS OF METABOLISM

ICIEM 2021

21-23 NOVEMBER 2021, SYDNEY, AUSTRALIA

An open-label single-centre pilot study to evaluate the safety and tolerability of pentosan polysulfate sodium in subjects with mucopolysaccharidosis type I (MPS I)

Audio File

Authors: Drago Bratkovic[1], Sharon Charles[2], David Ketteridge[1], Ravi Krishnan[2], Michael Imperiale[2]

Affiliations: [1]Women's and Children's Hospital, South Australia; [2]Paradigm Biopharmaceuticals Ltd., Melbourne, Australia

Introduction

- Patients with MPS I continue to experience pain and symptoms that affect function despite disease-modifying treatment with haematopoietic stem cell transplantation (HSCT) and enzyme replacement therapy (ERT)[1,2,3].
- Pentosan polysulfate sodium (PPS), a semisynthetic polysaccharide that resembles glycosaminoglycans (GAGs), has been shown to improve pain and function in patients with MPS I, MPS II and MPS VI [4, 5, 6].

Objectives

- Primary: Safety and tolerability of PPS.
- Secondary and exploratory: Patient reported outcomes (PROs) measuring pain, function, fatigue, quality of life (QOL); physical tests of mobility and dexterity; and pharmacokinetics (PK); changes in biomarkers signaling cartilage degeneration, bone turnover and inflammation.

PPS is being evaluated as an adjunctive therapy to ERT and/or HSCT in the PARA_MPS1_001 open-label, single-centre, phase 2 pilot study

Methods

Inclusion Criteria:

- Age ≥5 years, documented diagnosis of MPS I (genetic testing and/or enzyme activity), previously received HSCT and/or ERT.

Study Design:

- Patients are sequentially allocated to Cohort 1 or Cohort 2 and administered PPS subcutaneous (SC) injections once weekly to Week 12 then fortnightly to Week 48. Eligible patients may continue in the study for an additional 6 months.

PARA_MPSI_001 Study Design

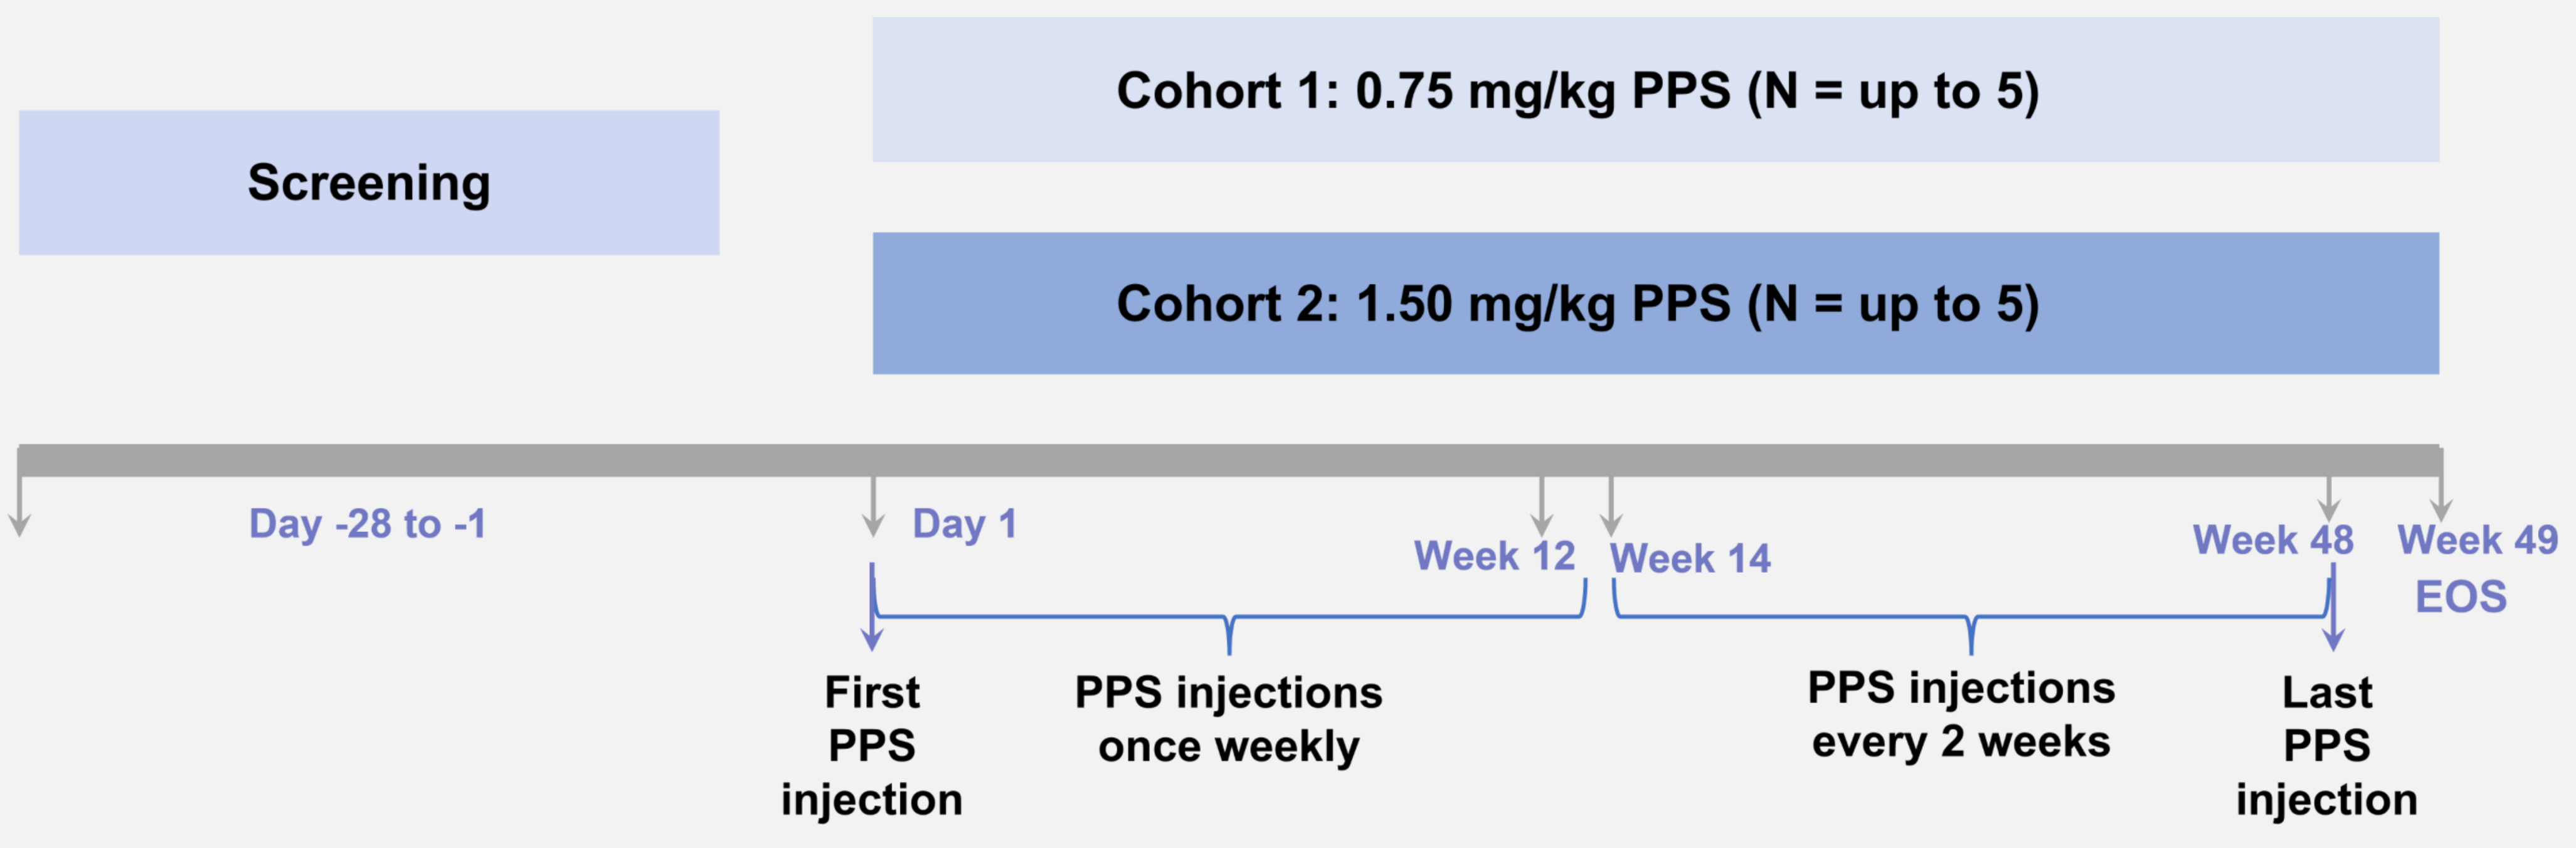
Cohort 1: 0.75 mg/kg PPS (N = up to 5)
Screening
Cohort 2: 1.50 mg/kg PPS (N = up to 5)
Day -28 to -1
Day 1
Week 12
Week 14
Week 48
Week 49
EOS
First PPS injection
PPS injections once weekly
PPS injections every 2 weeks
Last PPS injection

EOS = end of study; PPS = pentosan polysulfate sodium

Assessments:

- **Safety:** Incidence of treatment-emergent adverse events (TEAEs), serious adverse events (SAEs), and clinically significant changes from baseline in clinical laboratory data.
- **Efficacy:** Informed by global clinicians and an MPS patient focus group[7]:
 - **Physical tests:** 2- and 6-minute walk test (2/6MWT); 9-hole peg test (9HPT); Gait, Stairs, Gower, Chair (GSGC); joint passive range of motion (ROM) and grip test.
 - **PROs**: Paediatric, proxy, or adult Patient-Reported Outcomes Measurement Information System (PROMIS) - pain intensity, pain interference, pain behavior, fatigue, upper extremity and mobility/physical function with mobility aid short forms; and patient global impression of change (PGIC). Patients under 18 years had parent 'proxy' questionnaires completed as well as self-assessment.
- **PK:** PPS concentration in blood before and after repeat PPS administration (predose and 2, 4, and 6 hours post dose of dose 1, 4 and 18).
- **Biomarkers:** Urinary GAG (fragment and total GAG), and serum and urine biomarkers inflammation, cartilage degradation, and bone turnover.

PARA_MPSI_001 is ongoing; 3 of up to 10 planned patients are enrolled and receiving PPS. The 6-month data presented herein is for the 3 patients with a cut-off date of 6 August 2021.

Results

Patient Population and Disease Characteristics:

Three white, female, right-handed patients with MPS I-Hurler ages 14 (01-102), 15 (01-201) and 19 (01-101) years are enrolled into the study. All were diagnosed under 2 years of age and underwent HSCT within 6 months of diagnosis. Two patients (01-102, 01-201) received ERT for 6 months, commencing 4 and 2 months prior to HSCT respectively. Patients 01-101 and 01-102 received 0.75mg/kg, and patient 01-201 received 1.5mg/kg. All patients had musculoskeletal symptoms, pain, and reduced function at study entry. Analgesics were not permitted within 24 hours before assessments. Patient 01-101 had pre-planned bilateral knee surgery at week 15 and L) finger surgery at week 20.

Safety:

Of the 11 TEAEs, 2 were considered related to study drug (moderate pre-syncope and mild injection site bruising, both in the same patient and recovered). All TEAEs have been deemed mild to moderate and resolved, with no SAEs reported.

Biomarkers:

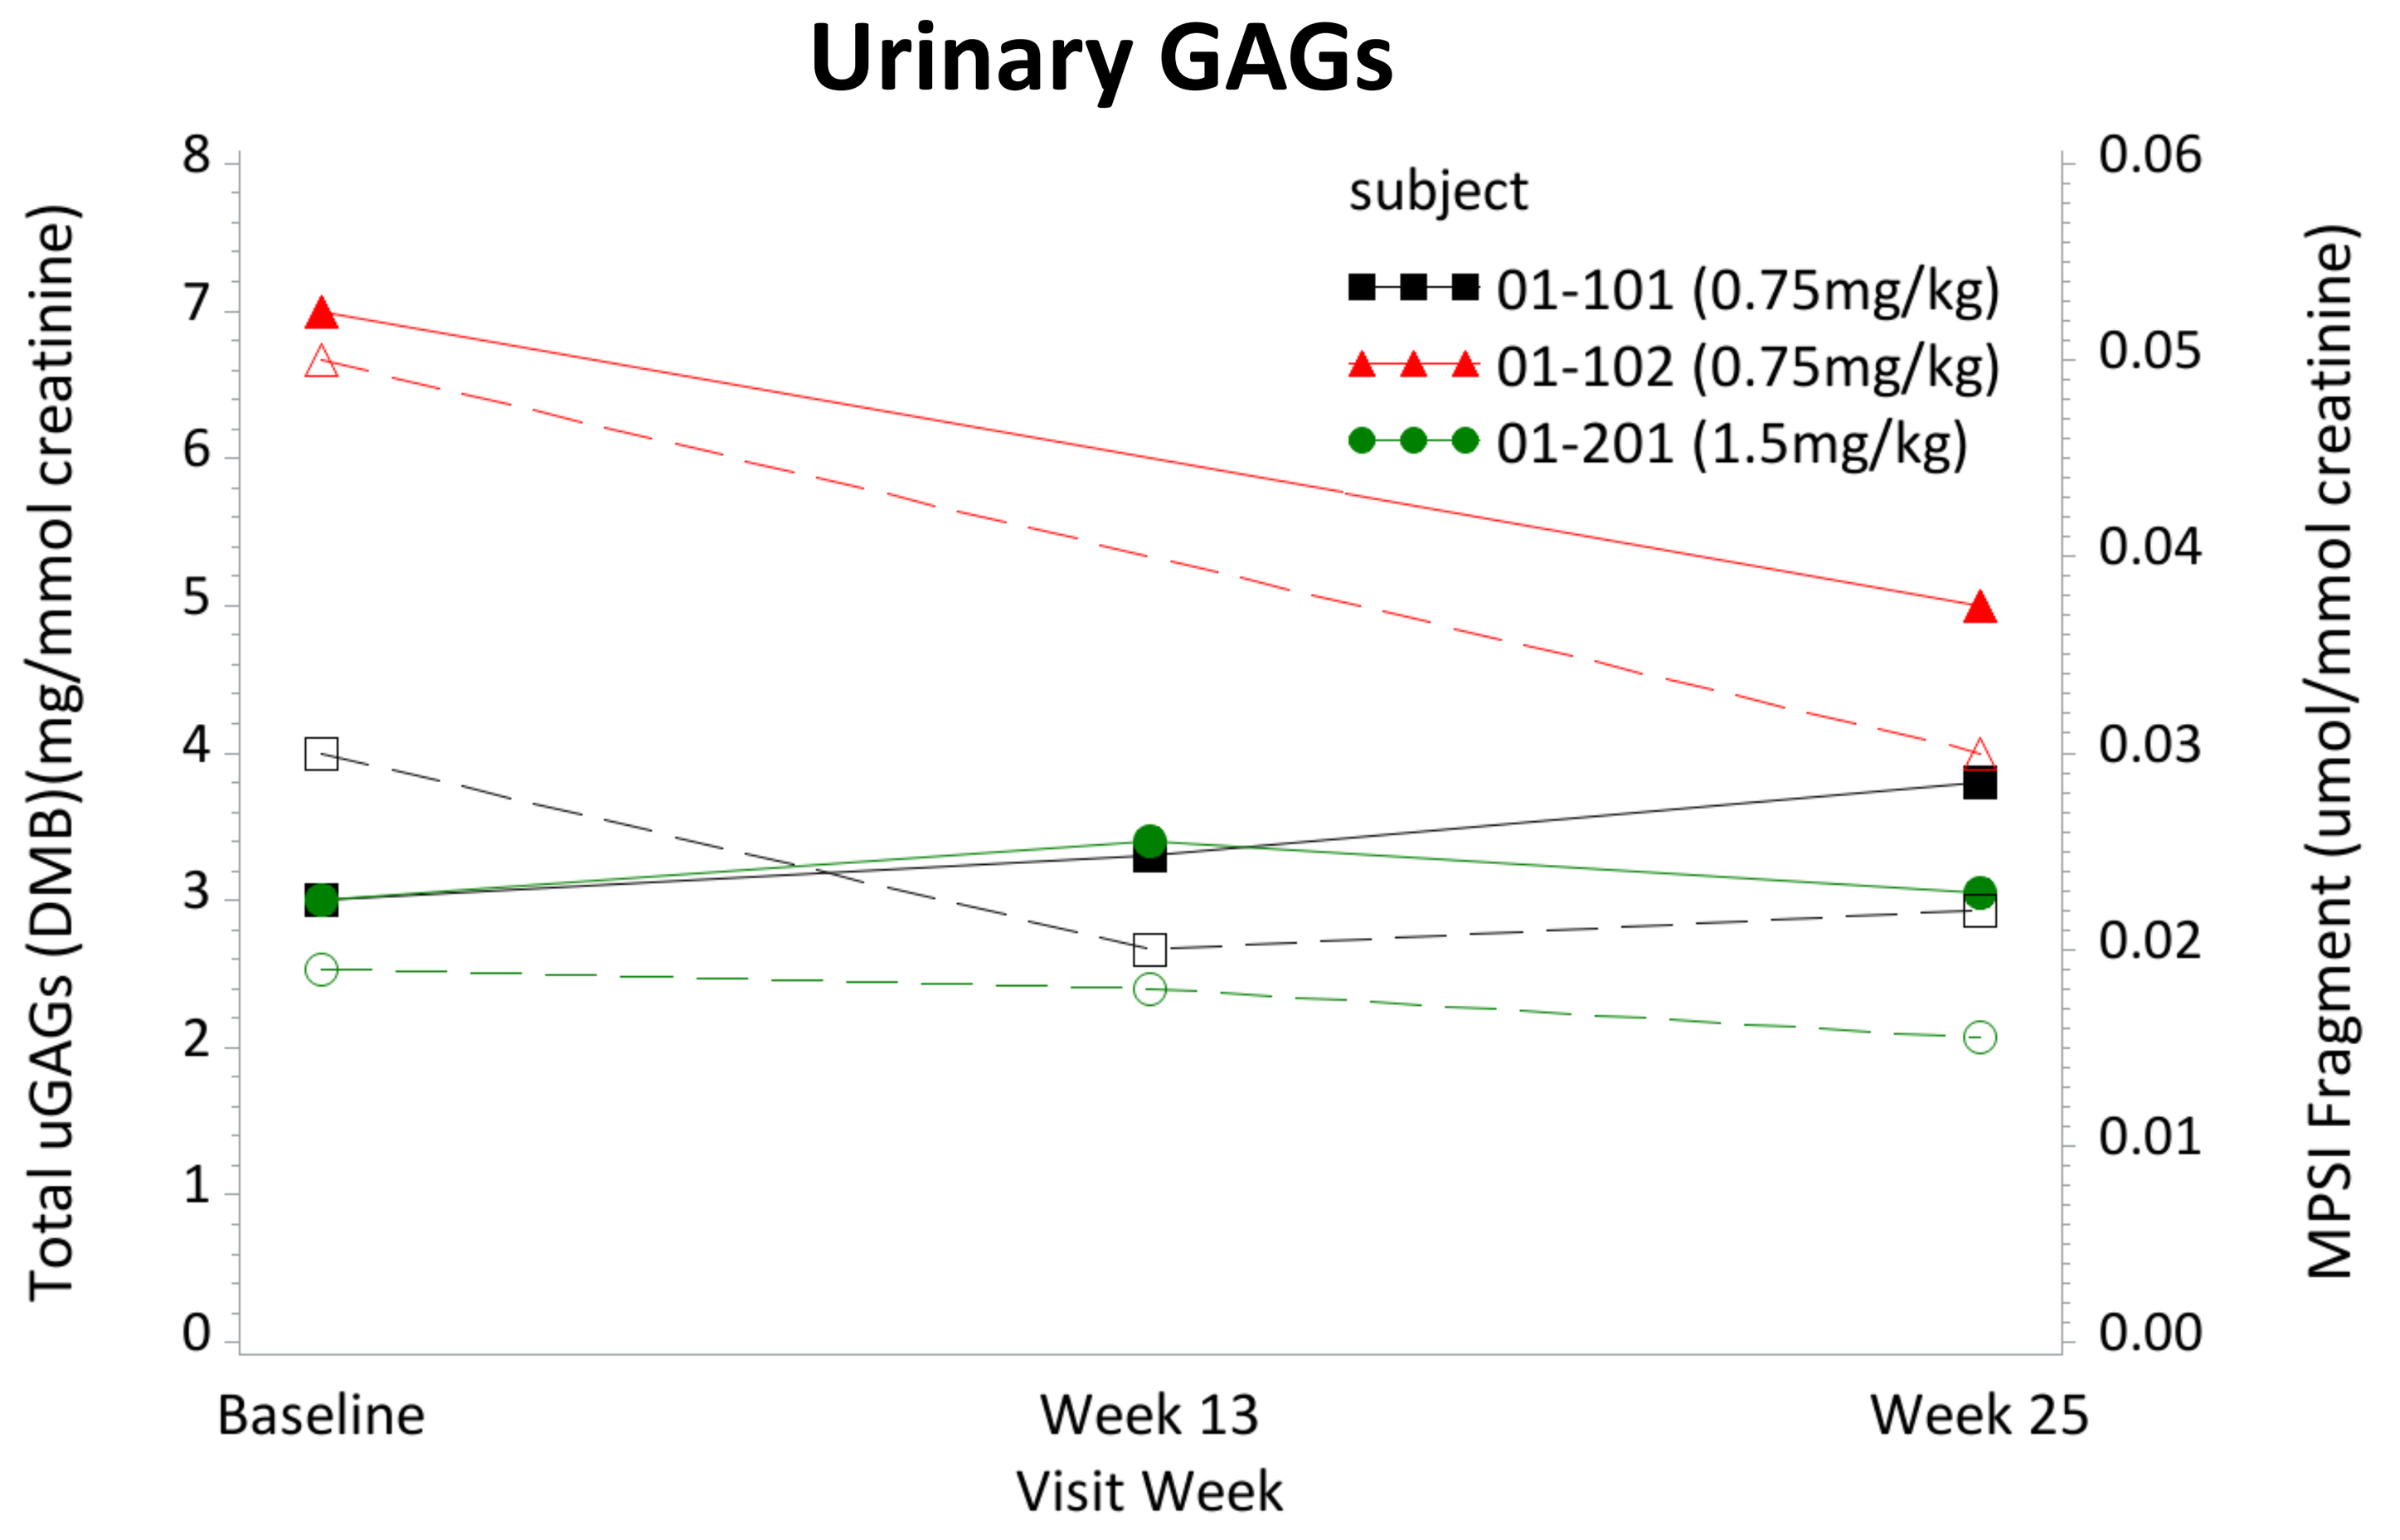
Urinary GAGs
subject
01-101 (0.75mg/kg)
01-102 (0.75mg/kg)
01-201 (1.5mg/kg)
Total uGAGs (DMB)(mg/mmol creatinine)
MPSI Fragment (umol/mmol creatinine)
0
1
2
3
4
5
6
7
8
0.00
0.01
0.02
0.03
0.04
0.05
0.06
Baseline
Week 13
Week 25
Visit Week

____Solid lines represent Total GAGs and . . .dotted lines represent MPSI Fragment

- All patients had a decrease (improvement) in the MPS I signature oligosaccharide GAG fragments (sulfated disaccharide) (UA-HNAc S)[8] from baseline to week 25, with the greatest improvement seen in patient 01-102, who had a 40% drop from the highest baseline levels.
- There was a modest increase in total uGAGs (dimethylene blue (DMB)) for 2 patients, and a decrease in patient 01-102.

Results (cont.)

Pharmacokinetics:

Plasma PPS concentration vs. time profiles from patients with MPS1, grouped by study day

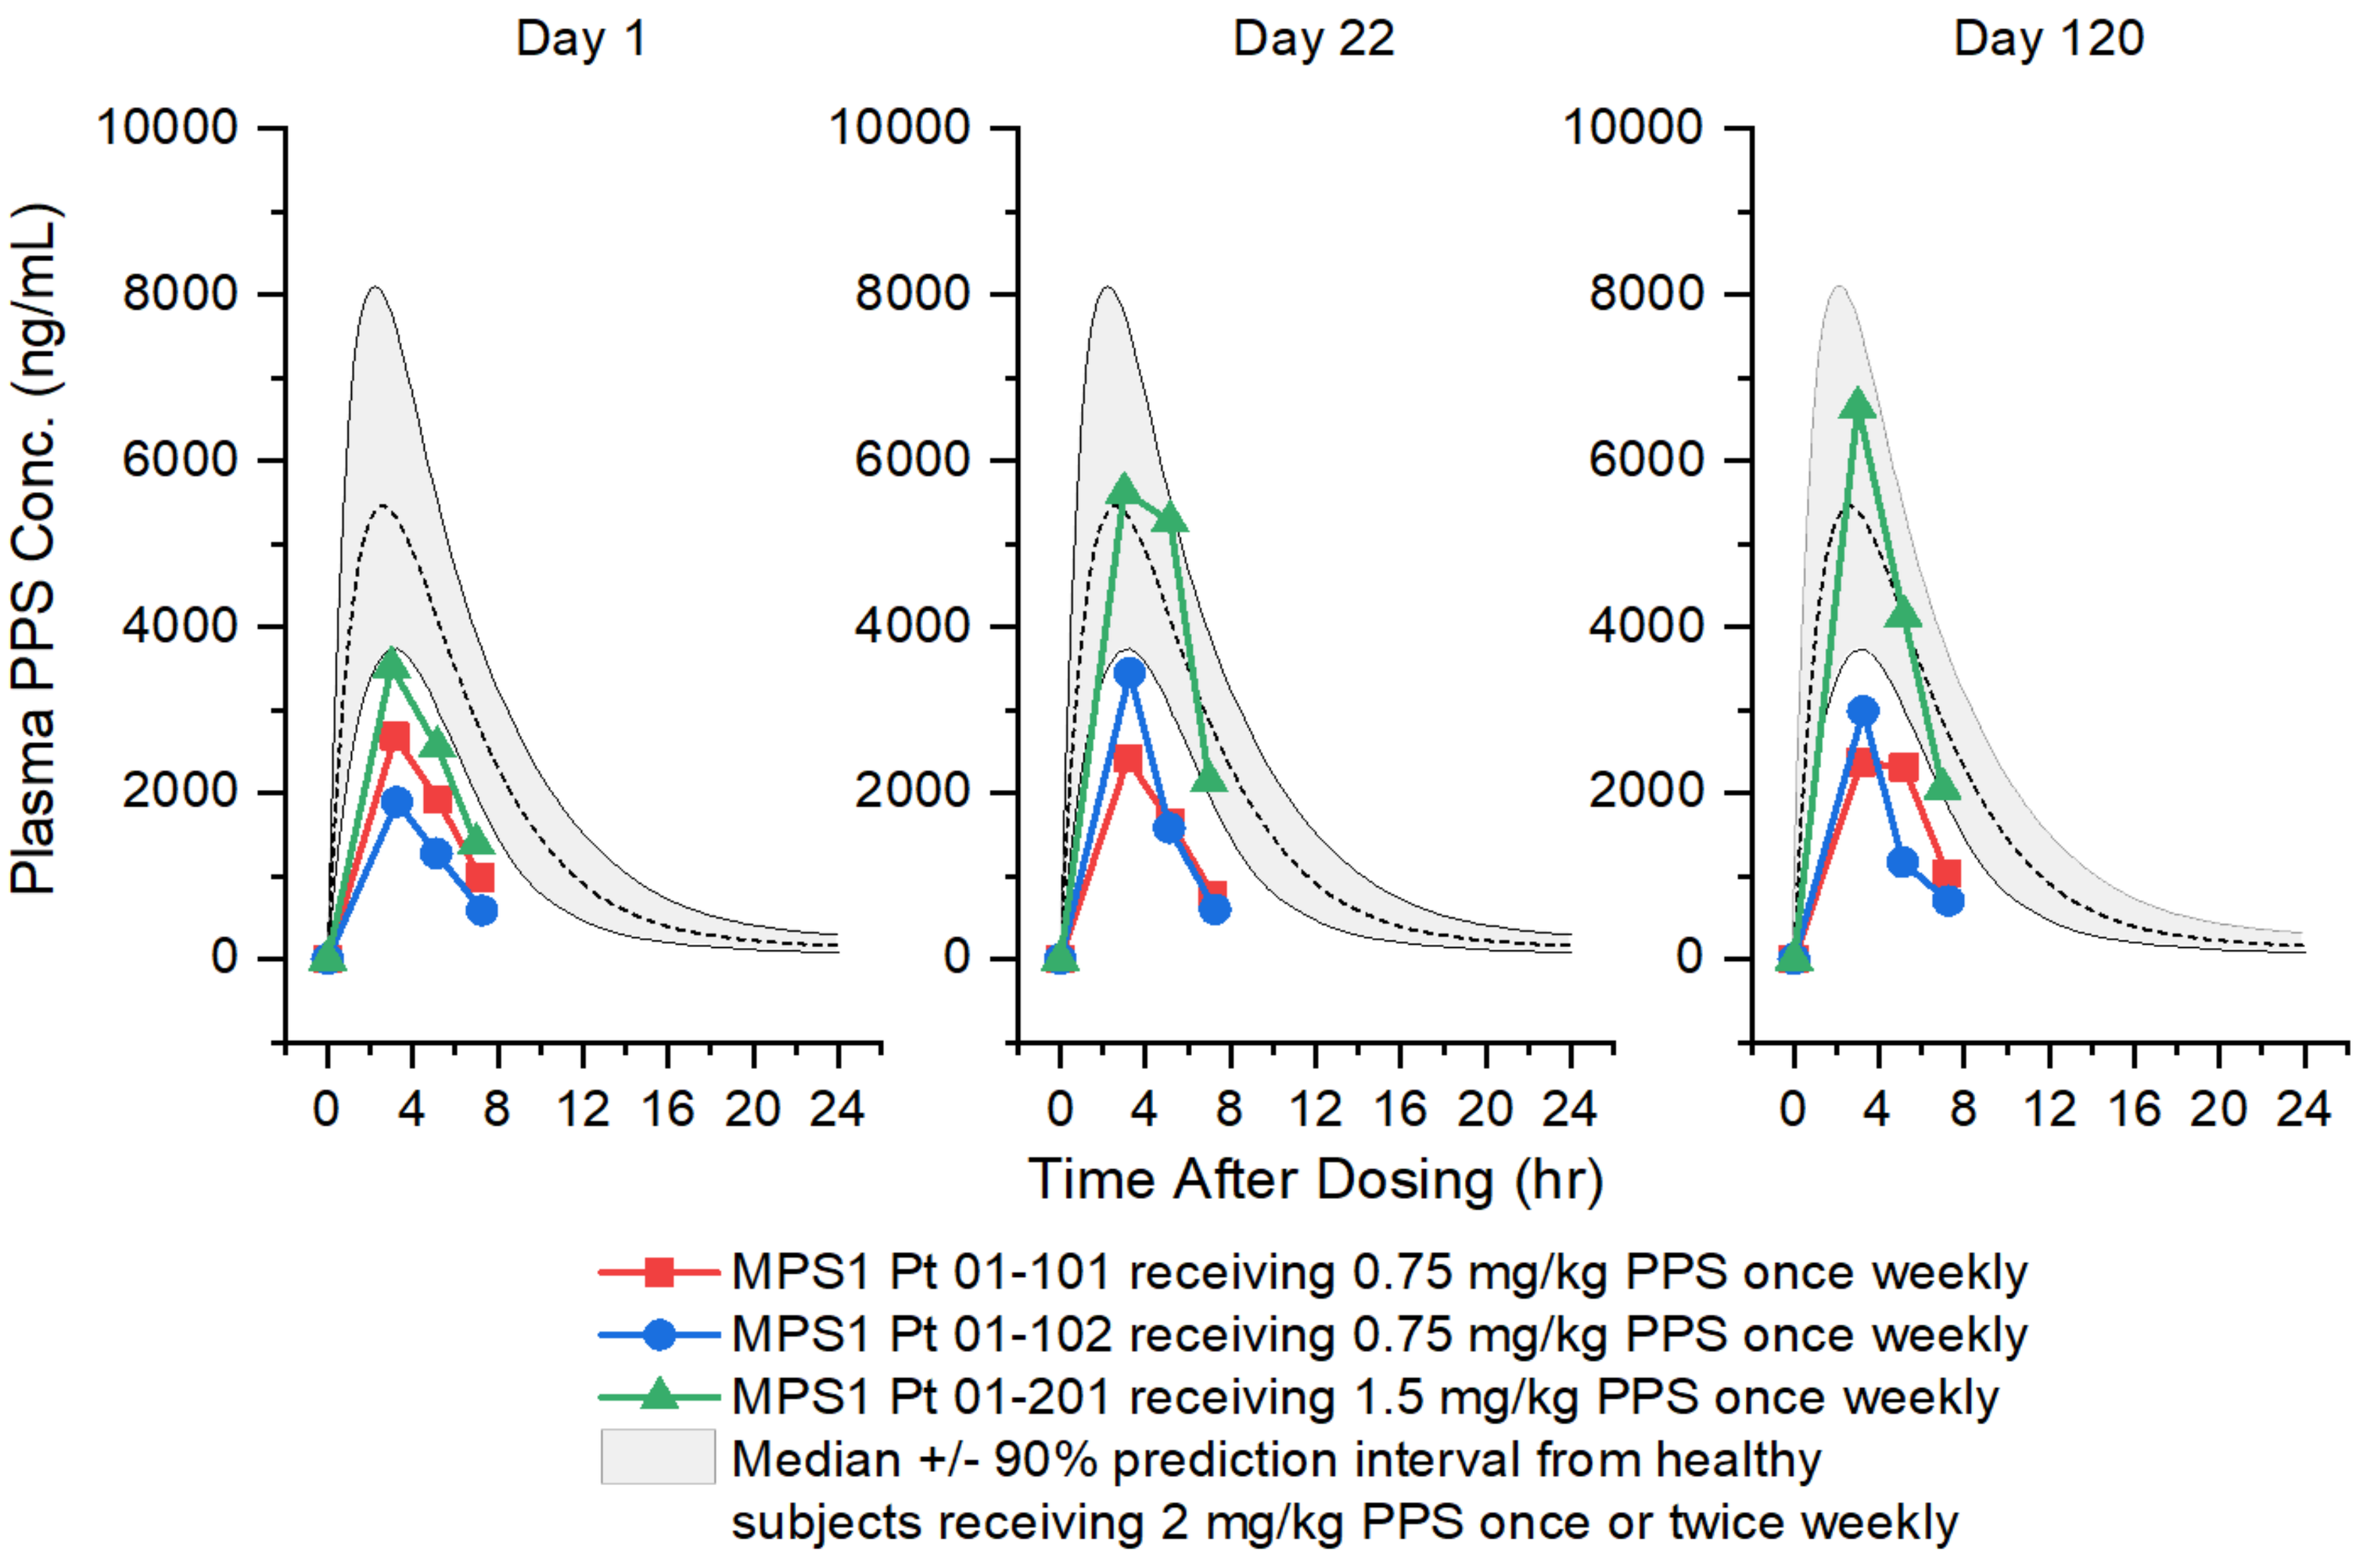
Day 1
Day 22
Day 120
Plasma PPS Conc. (ng/mL)
10000
8000
6000
4000
2000
0
0 4 8 12 16 20 24
Time After Dosing (hr)
MPS1 Pt 01-101 receiving 0.75 mg/kg PPS once weekly
MPS1 Pt 01-102 receiving 0.75 mg/kg PPS once weekly
MPS1 Pt 01-201 receiving 1.5 mg/kg PPS once weekly
Median +/- 90% prediction interval from healthy subjects receiving 2 mg/kg PPS once or twice weekly

- The PK characteristics were reproducible, demonstrated consistent rapid rise and fall in plasma concentration that was proportionate to dose administered (mg/kg). Plasma concentration returned to baseline prior to the next dose / assessment and followed characteristics of repeated single dose exposure.

Biomarkers:

Cartilage Degeneration and Bone Turnover

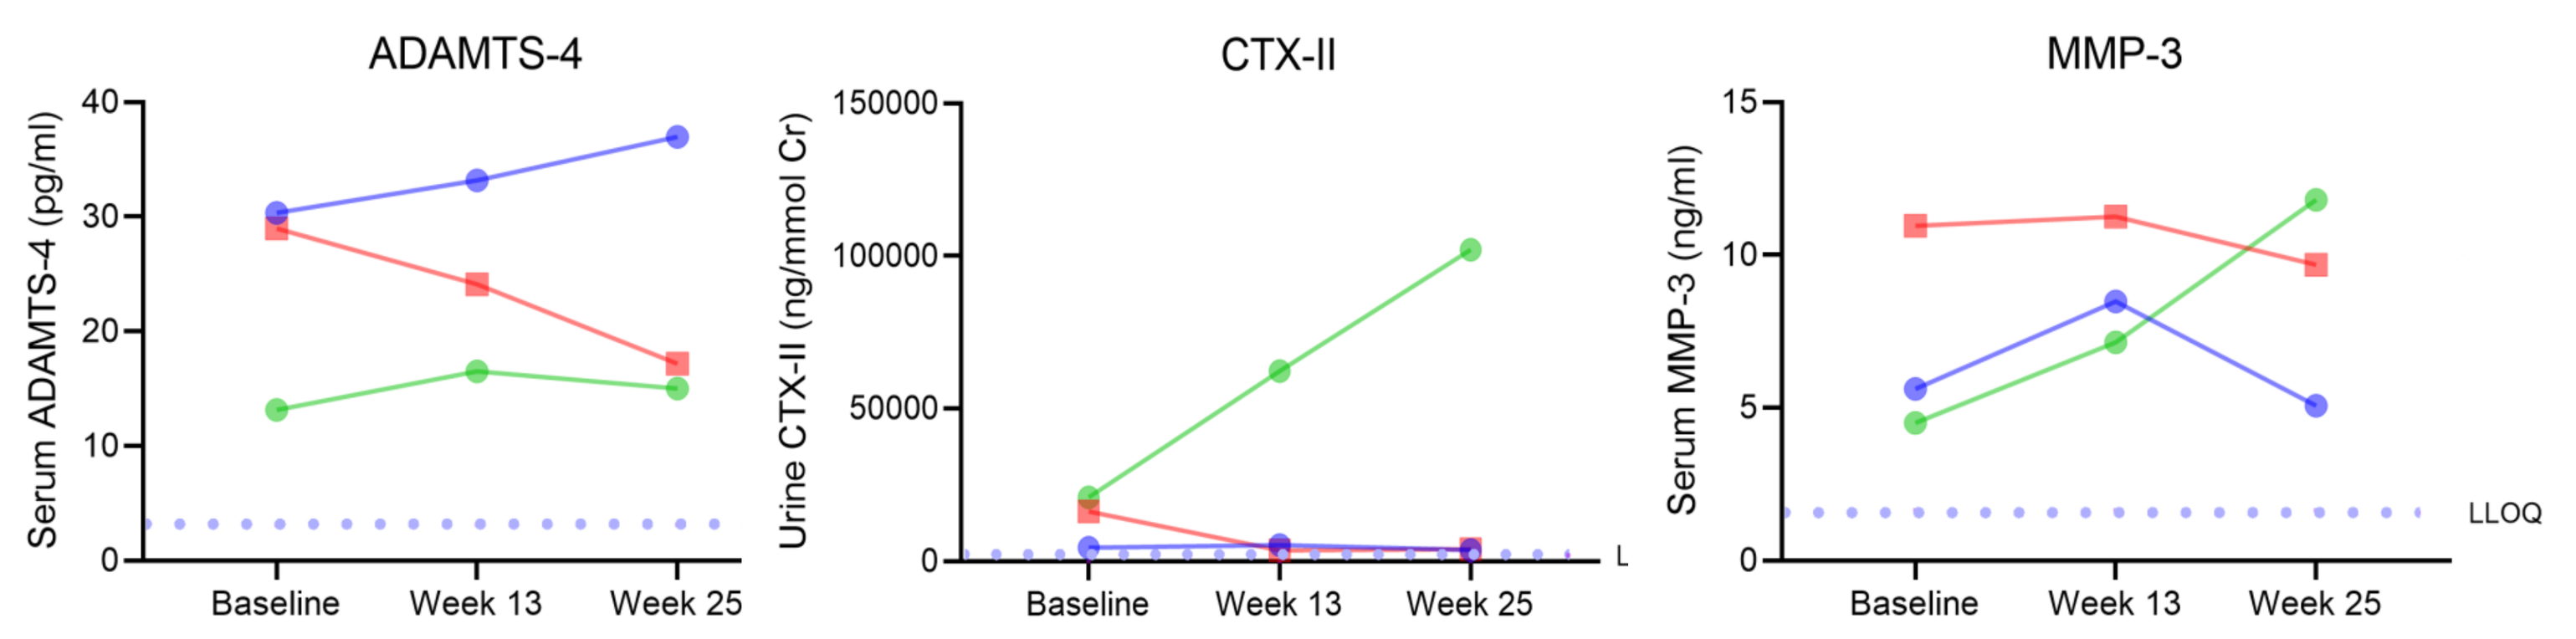
ADAMTS-4
Serum ADAMTS-4 (pg/ml)
CTX-II
Urine CTX-II (ng/mmol Cr)
MMP-3
Serum MMP-3 (ng/ml)
LLOQ
Baseline
Week 13
Week 25

Inflammation

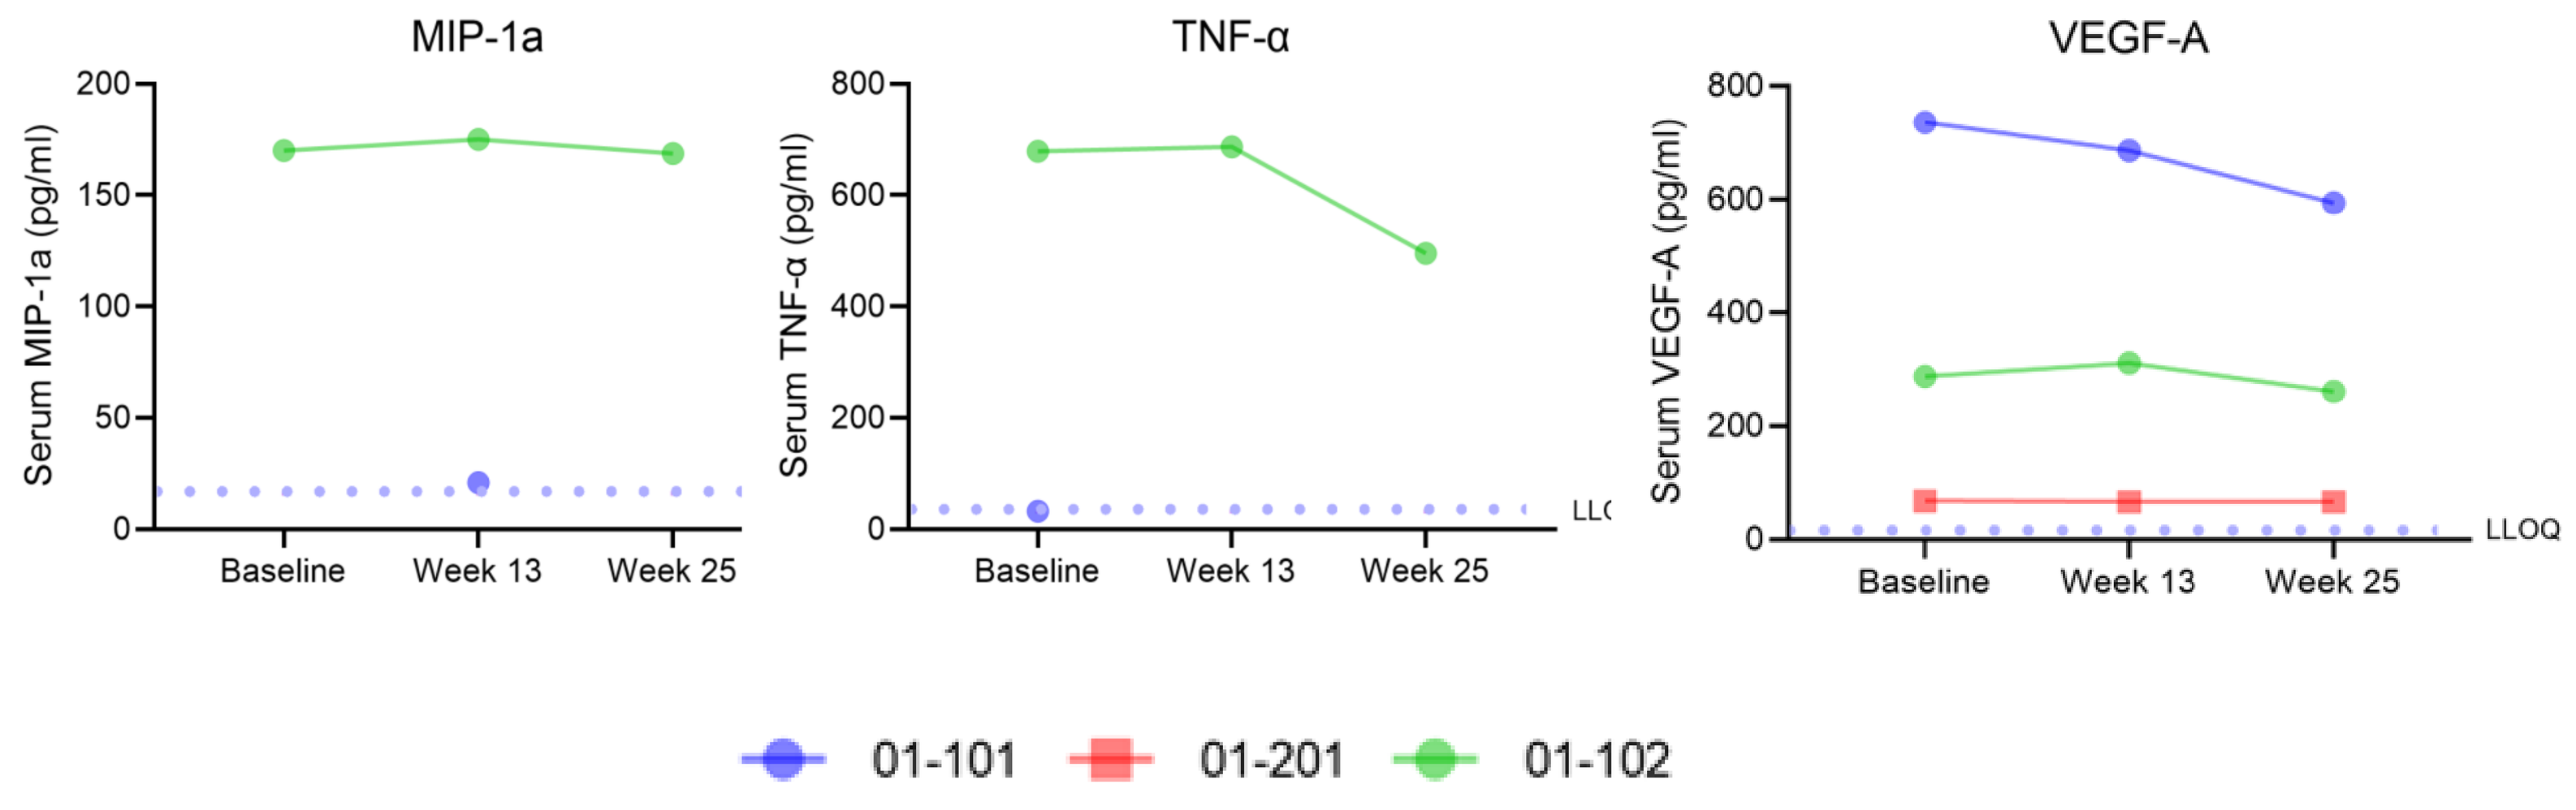
MIP-1a
Serum MIP-1a (pg/ml)
TNF-α
Serum TNF-α (pg/ml)
VEGF-A
Serum VEGF-A (pg/ml)
LLOQ
Baseline
Week 13
Week 25
01-101
01-201
01-102

- Overall, most of the biomarkers (inflammatory, cartilage degeneration and bone turnover) evaluated in all patients have indicated either no change or a reduction in levels at week 25 compared to baseline. This is an indication that PPS has the potential to modulate the inflammatory and joint degenerating biomarkers that are associated with arthralgia in MPS I patients.

Efficacy:

2/6MWT

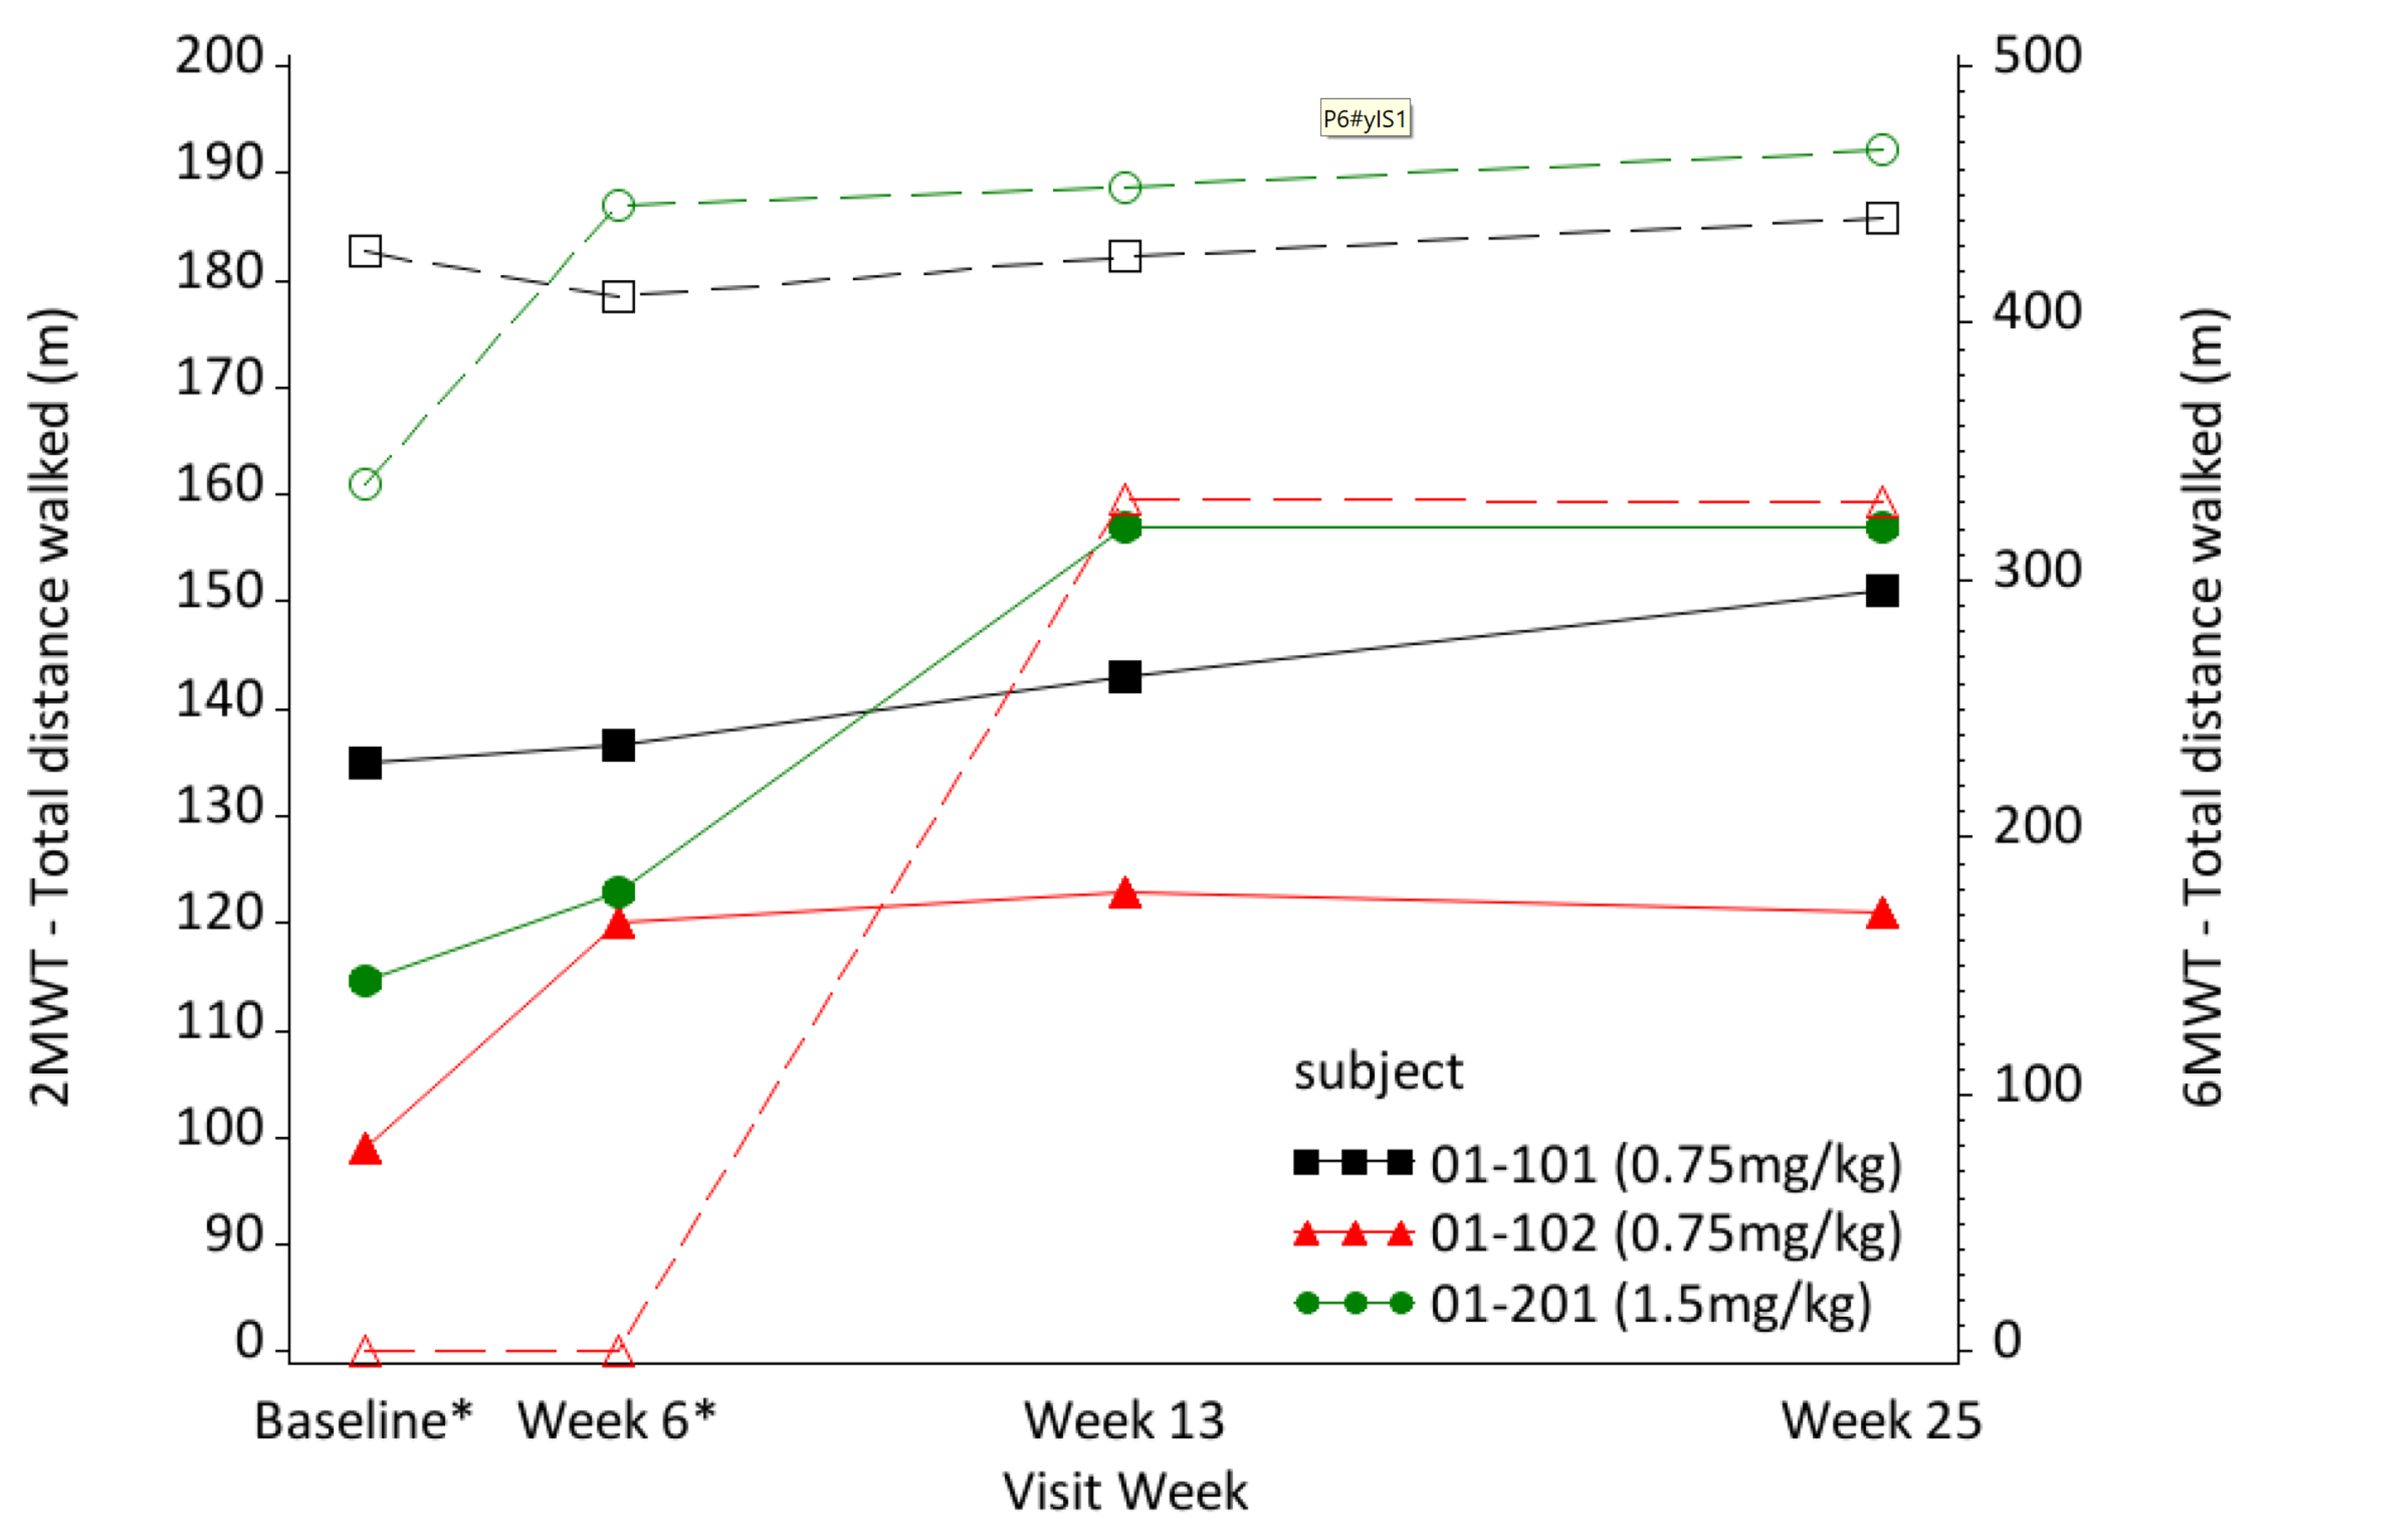
2MWT - Total distance walked (m)
6MWT - Total distance walked (m)
P6#yIS1
subject
01-101 (0.75mg/kg)
01-102 (0.75mg/kg)
01-201 (1.5mg/kg)
Baseline*
Week 6*
Week 13
Week 25
Visit Week

*Subject 01-102 was unable to complete the 6MWT at baseline due to pain and at week 6 due to fatigue. ____solid lines represent 2MWT and . . . dotted lines represent 6MWT

- There was a trend toward improvement in 2/6MWT from baseline to week 25. Higher functioning patients at baseline demonstrated modest improvement.
- 01-102 was unable to complete the full 6MWT at baseline and week 6 due to back pain and fatigue (baseline), and fatigue (W6), however was able to complete the full 6 minutes at subsequent visits.

ROM

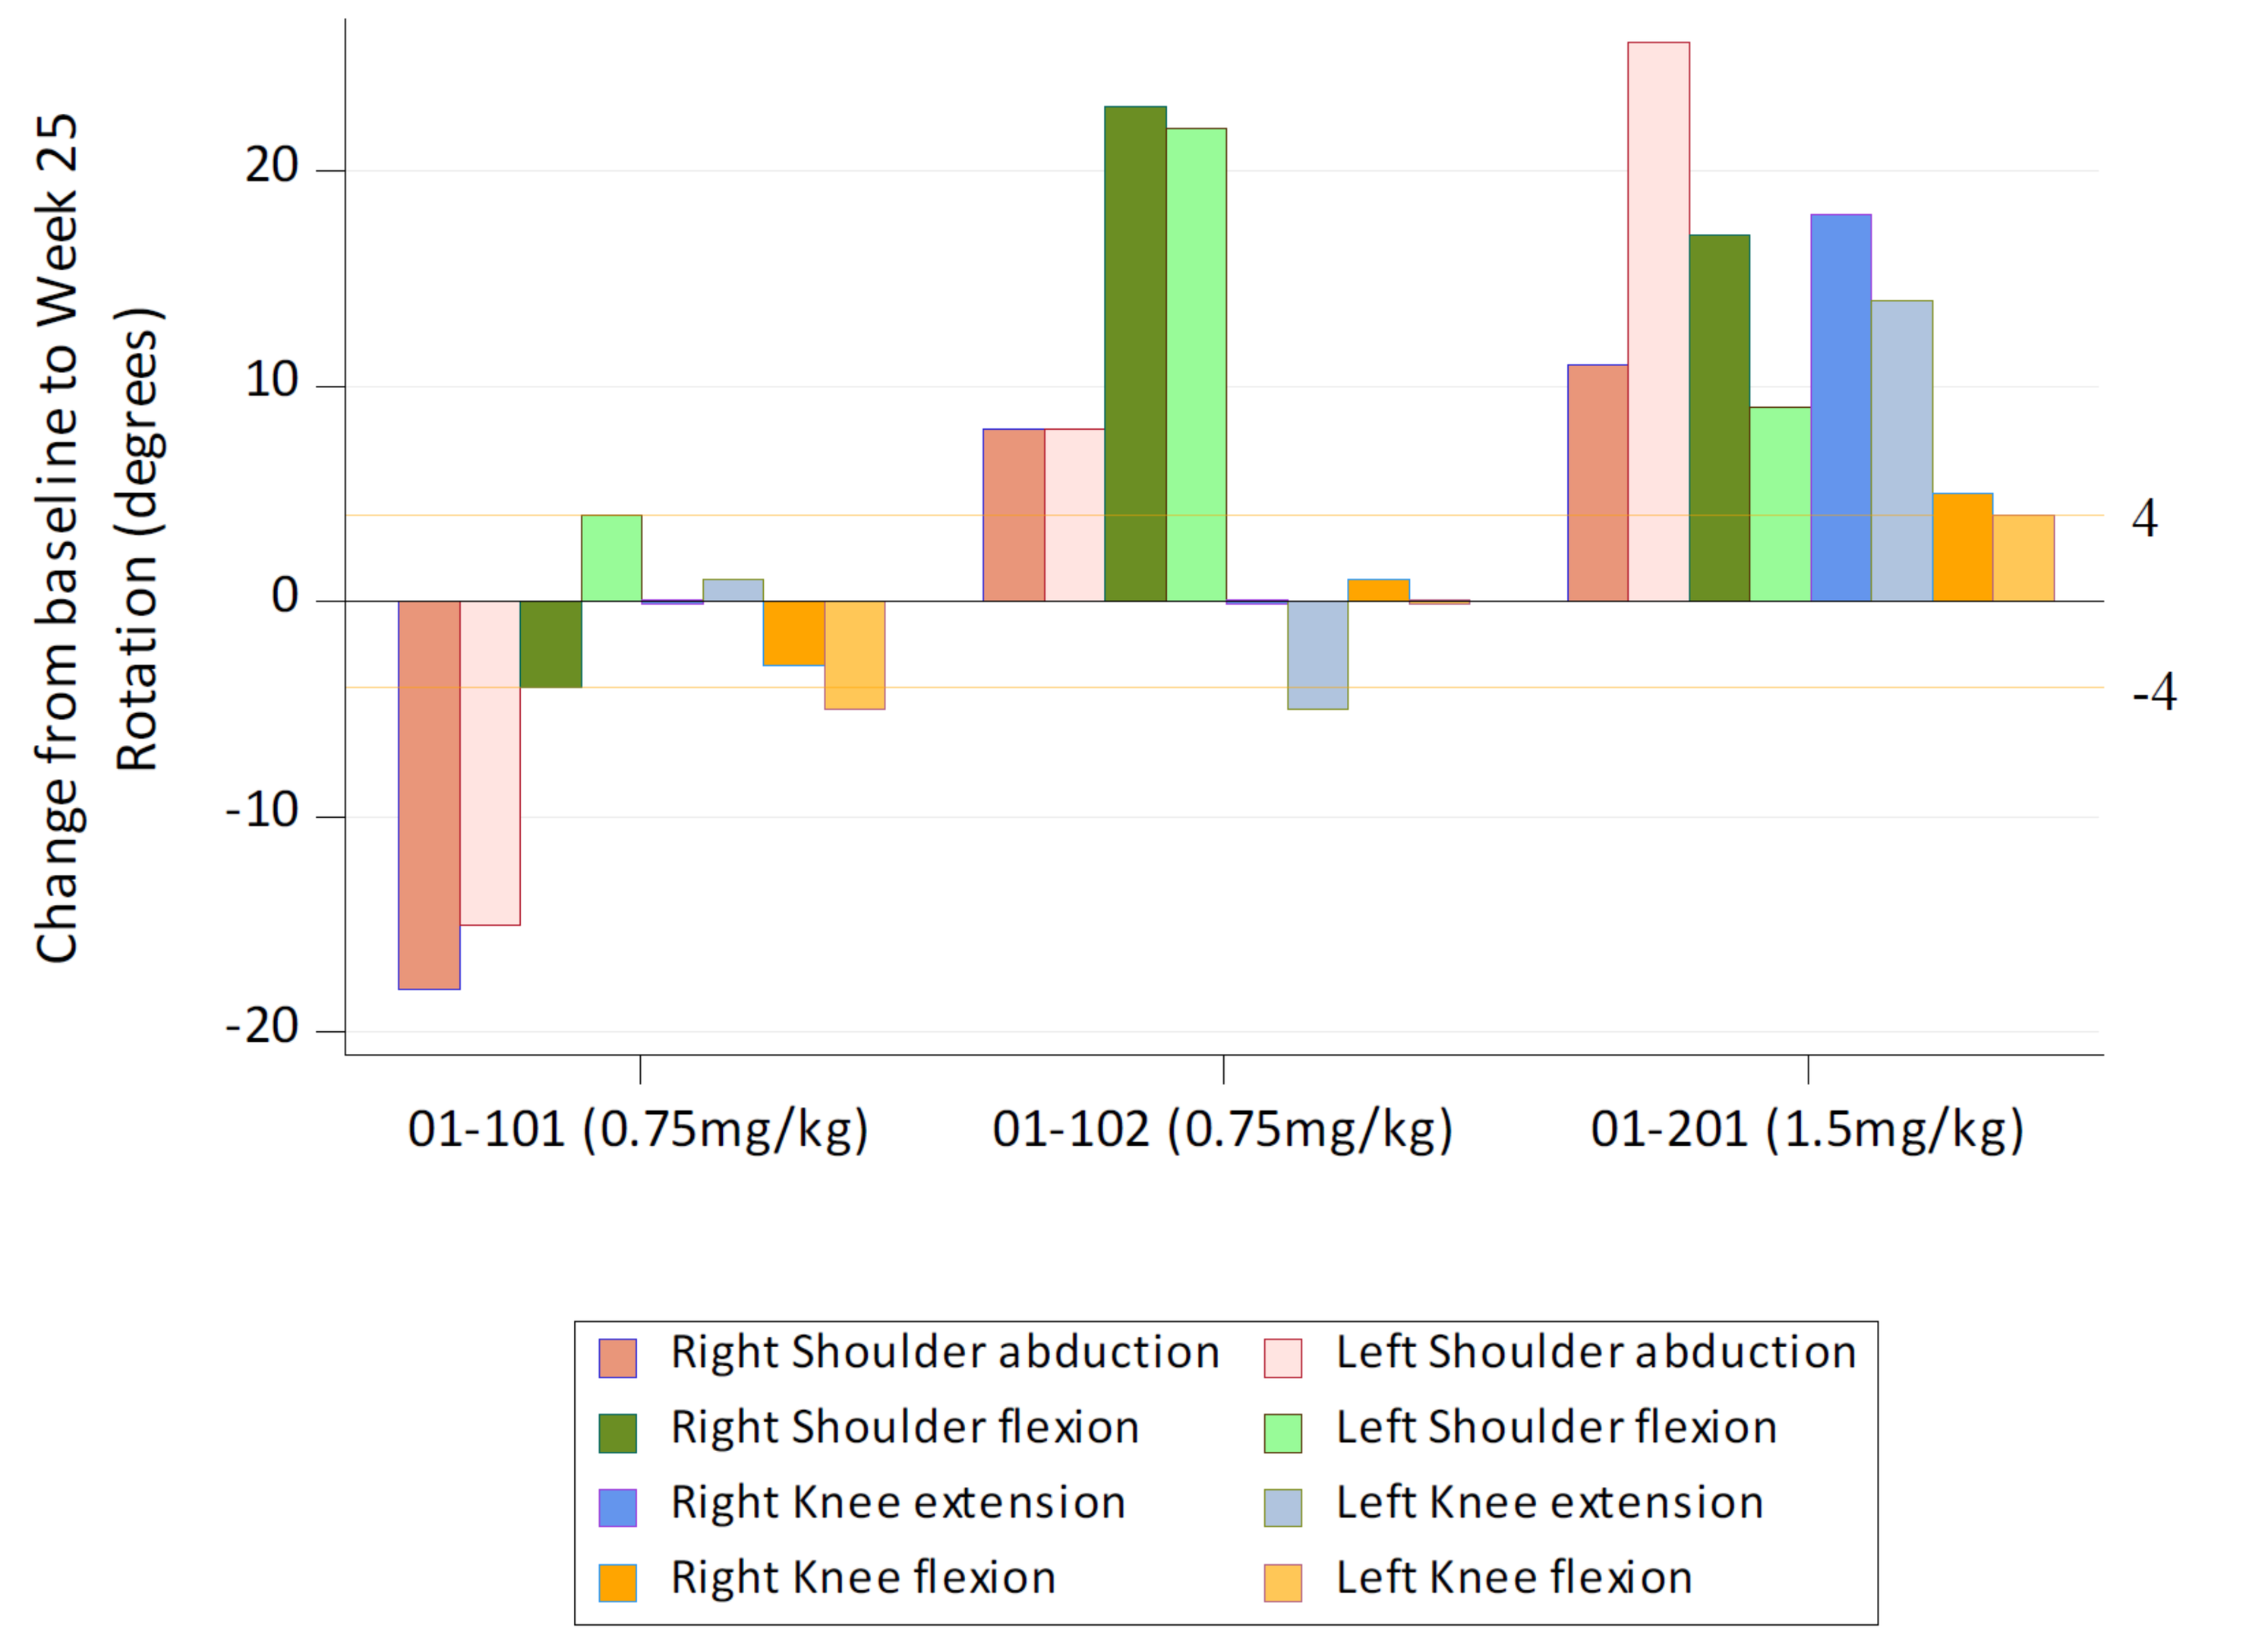
Change from baseline to Week 25
Rotation (degrees)
01-101 (0.75mg/kg)
01-102 (0.75mg/kg)
01-201 (1.5mg/kg)
4
-4
Right Shoulder abduction
Left Shoulder abduction
Right Shoulder flexion
Left Shoulder flexion
Right Knee extension
Left Knee extension
Right Knee flexion
Left Knee flexion

ROM measurements are considered no change if they are within +/- 4 degrees change from baseline[4]

- At week 25, patients exhibited either improvement or no significant change on most parameters. Exception was 01-101, who had greater shoulder abduction ROM at baseline than the other patients, and experienced decreased shoulder abduction movement at week 25. Knee extension remained unchanged in 2 of the 3 patients, and significantly improved in the third patient. Patient 01-201, receiving the high dose (1.5 mg/kg) had improved ROM on all joints assessed.

9-Hole Peg Test

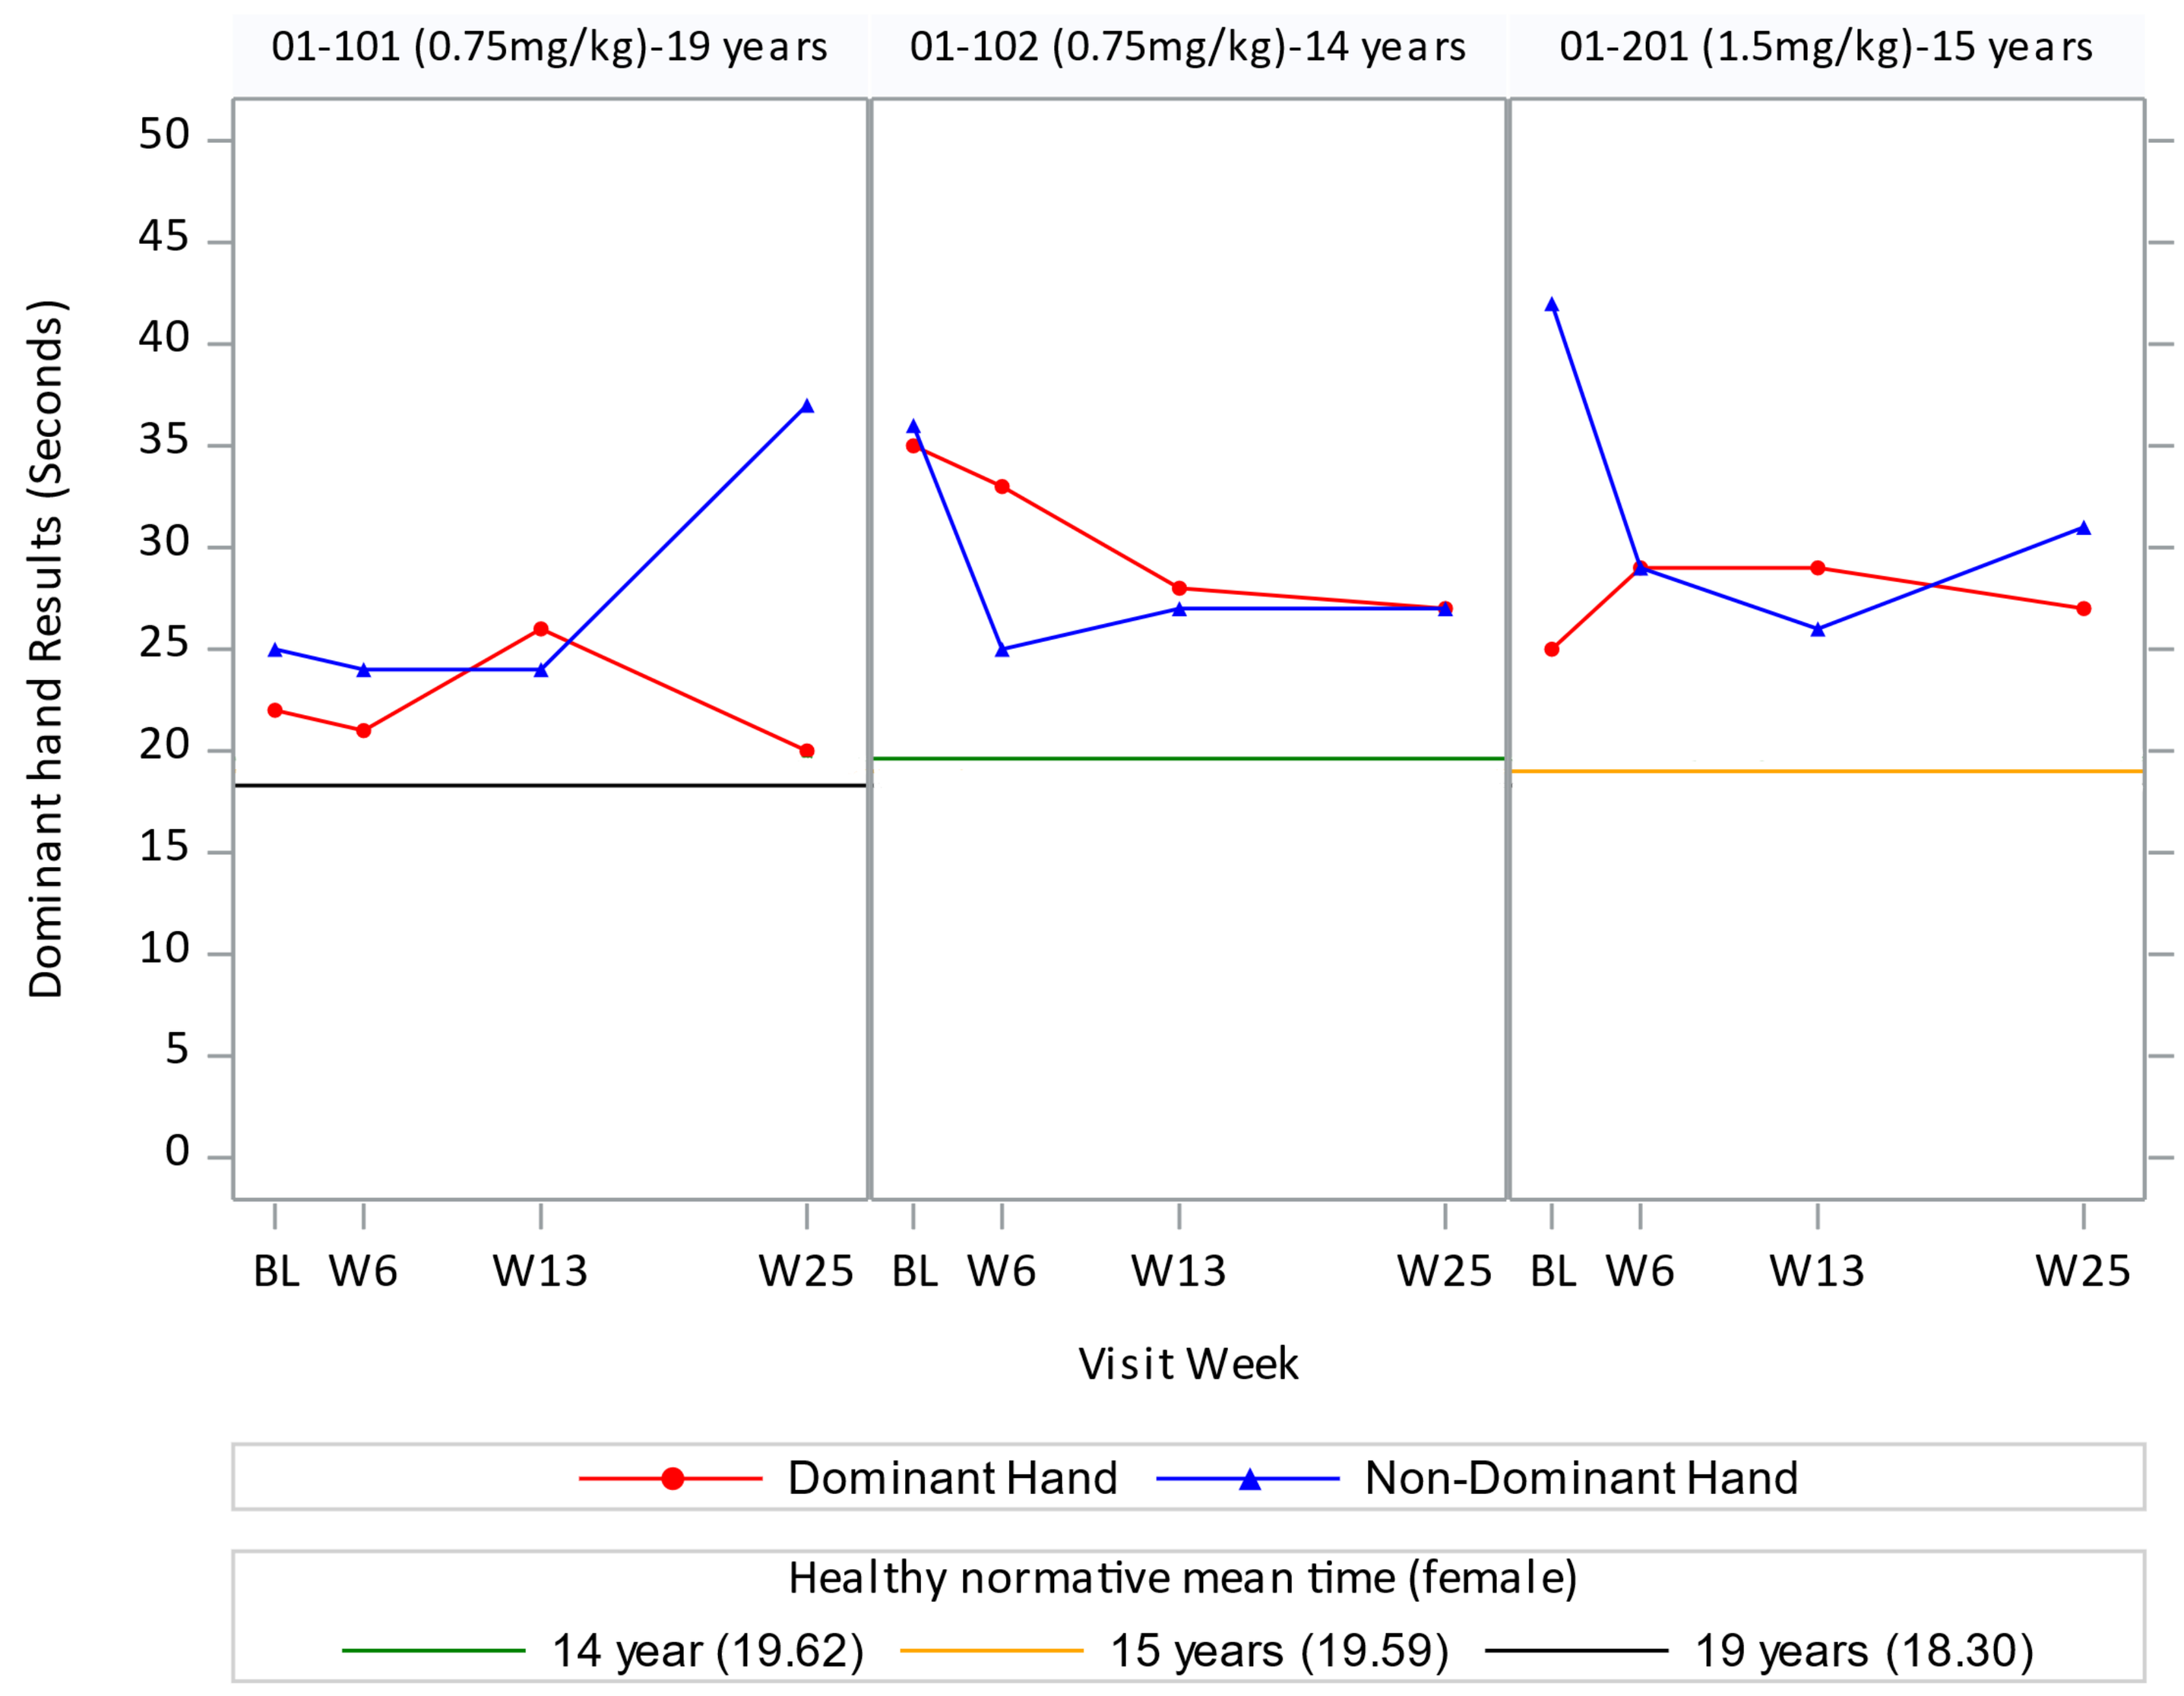
01-101 (0.75mg/kg)-19 years
01-102 (0.75mg/kg)-14 years
01-201 (1.5mg/kg)-15 years
Dominant hand Results (Seconds)
Non-Dominant hand Results (Seconds)
BL W6 W13 W25
Visit Week
Dominant Hand
Non-Dominant Hand
Healthy normative mean time (female)
14 year (19.62)
15 years (19.59)
19 years (18.30)

- At baseline, all patients took longer to complete the test than the average for age/gender, with two patients taking greater than 2 standard deviations longer. At week 25, patients were generally stable or improved in the test (exception was patient 01-101 non-dominant hand, possibly influenced by L) index finger surgery at week 20).

Gait, Stair, Gowers, Chair (GSGC)

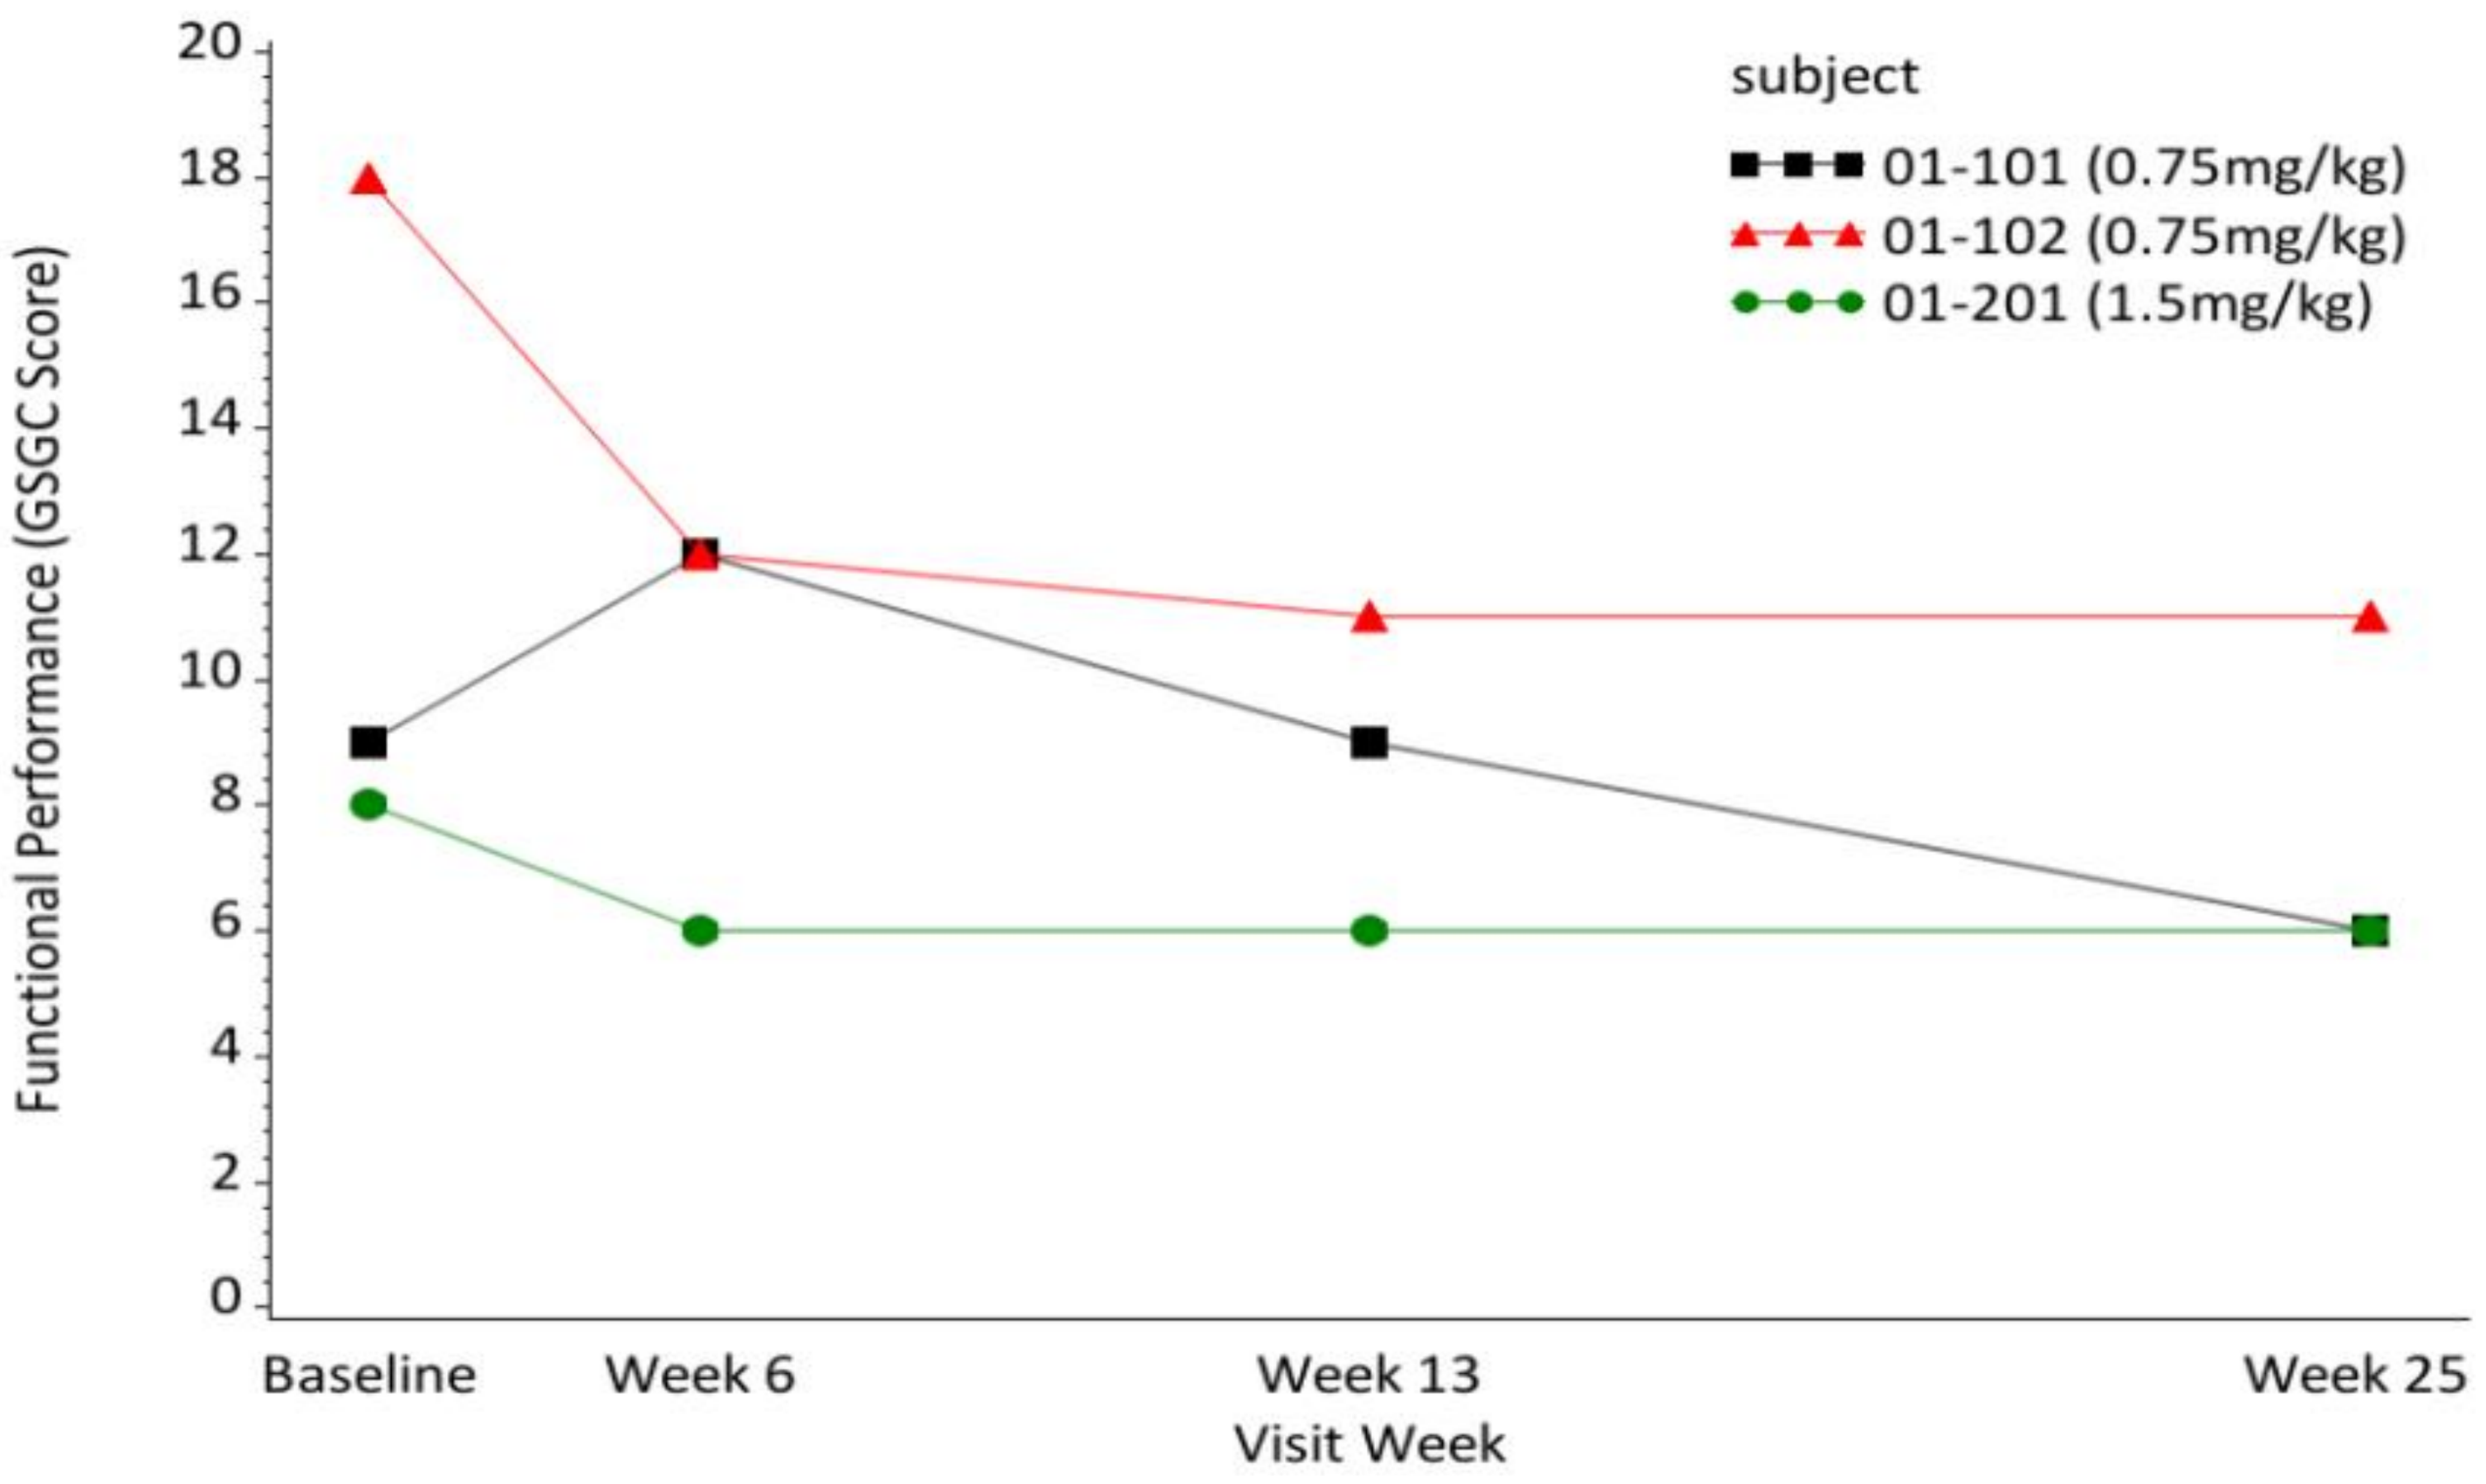
Functional Performance (GSGC Score)
subject
01-101 (0.75mg/kg)
01-102 (0.75mg/kg)
01-201 (1.5mg/kg)
Baseline
Week 6
Week 13
Week 25
Visit Week

Decreasing score is improvement and increasing score is worsening

- All patients demonstrated overall improvement in the GSGC qualitative global score with improvements ranging from 25%-39% over baseline at week 25.
- Times to complete the tests were faster at week 25 than baseline, with the exception was 01-101 (who had a faster time at study entry, and had bilateral knee surgery at week 15), and increased seconds in Gower and chair activities at week 25 (data not shown).

Grip strength: No significant changes in were observed from baseline to week 25.

Results (cont.)

PROMIS Pain Intensity

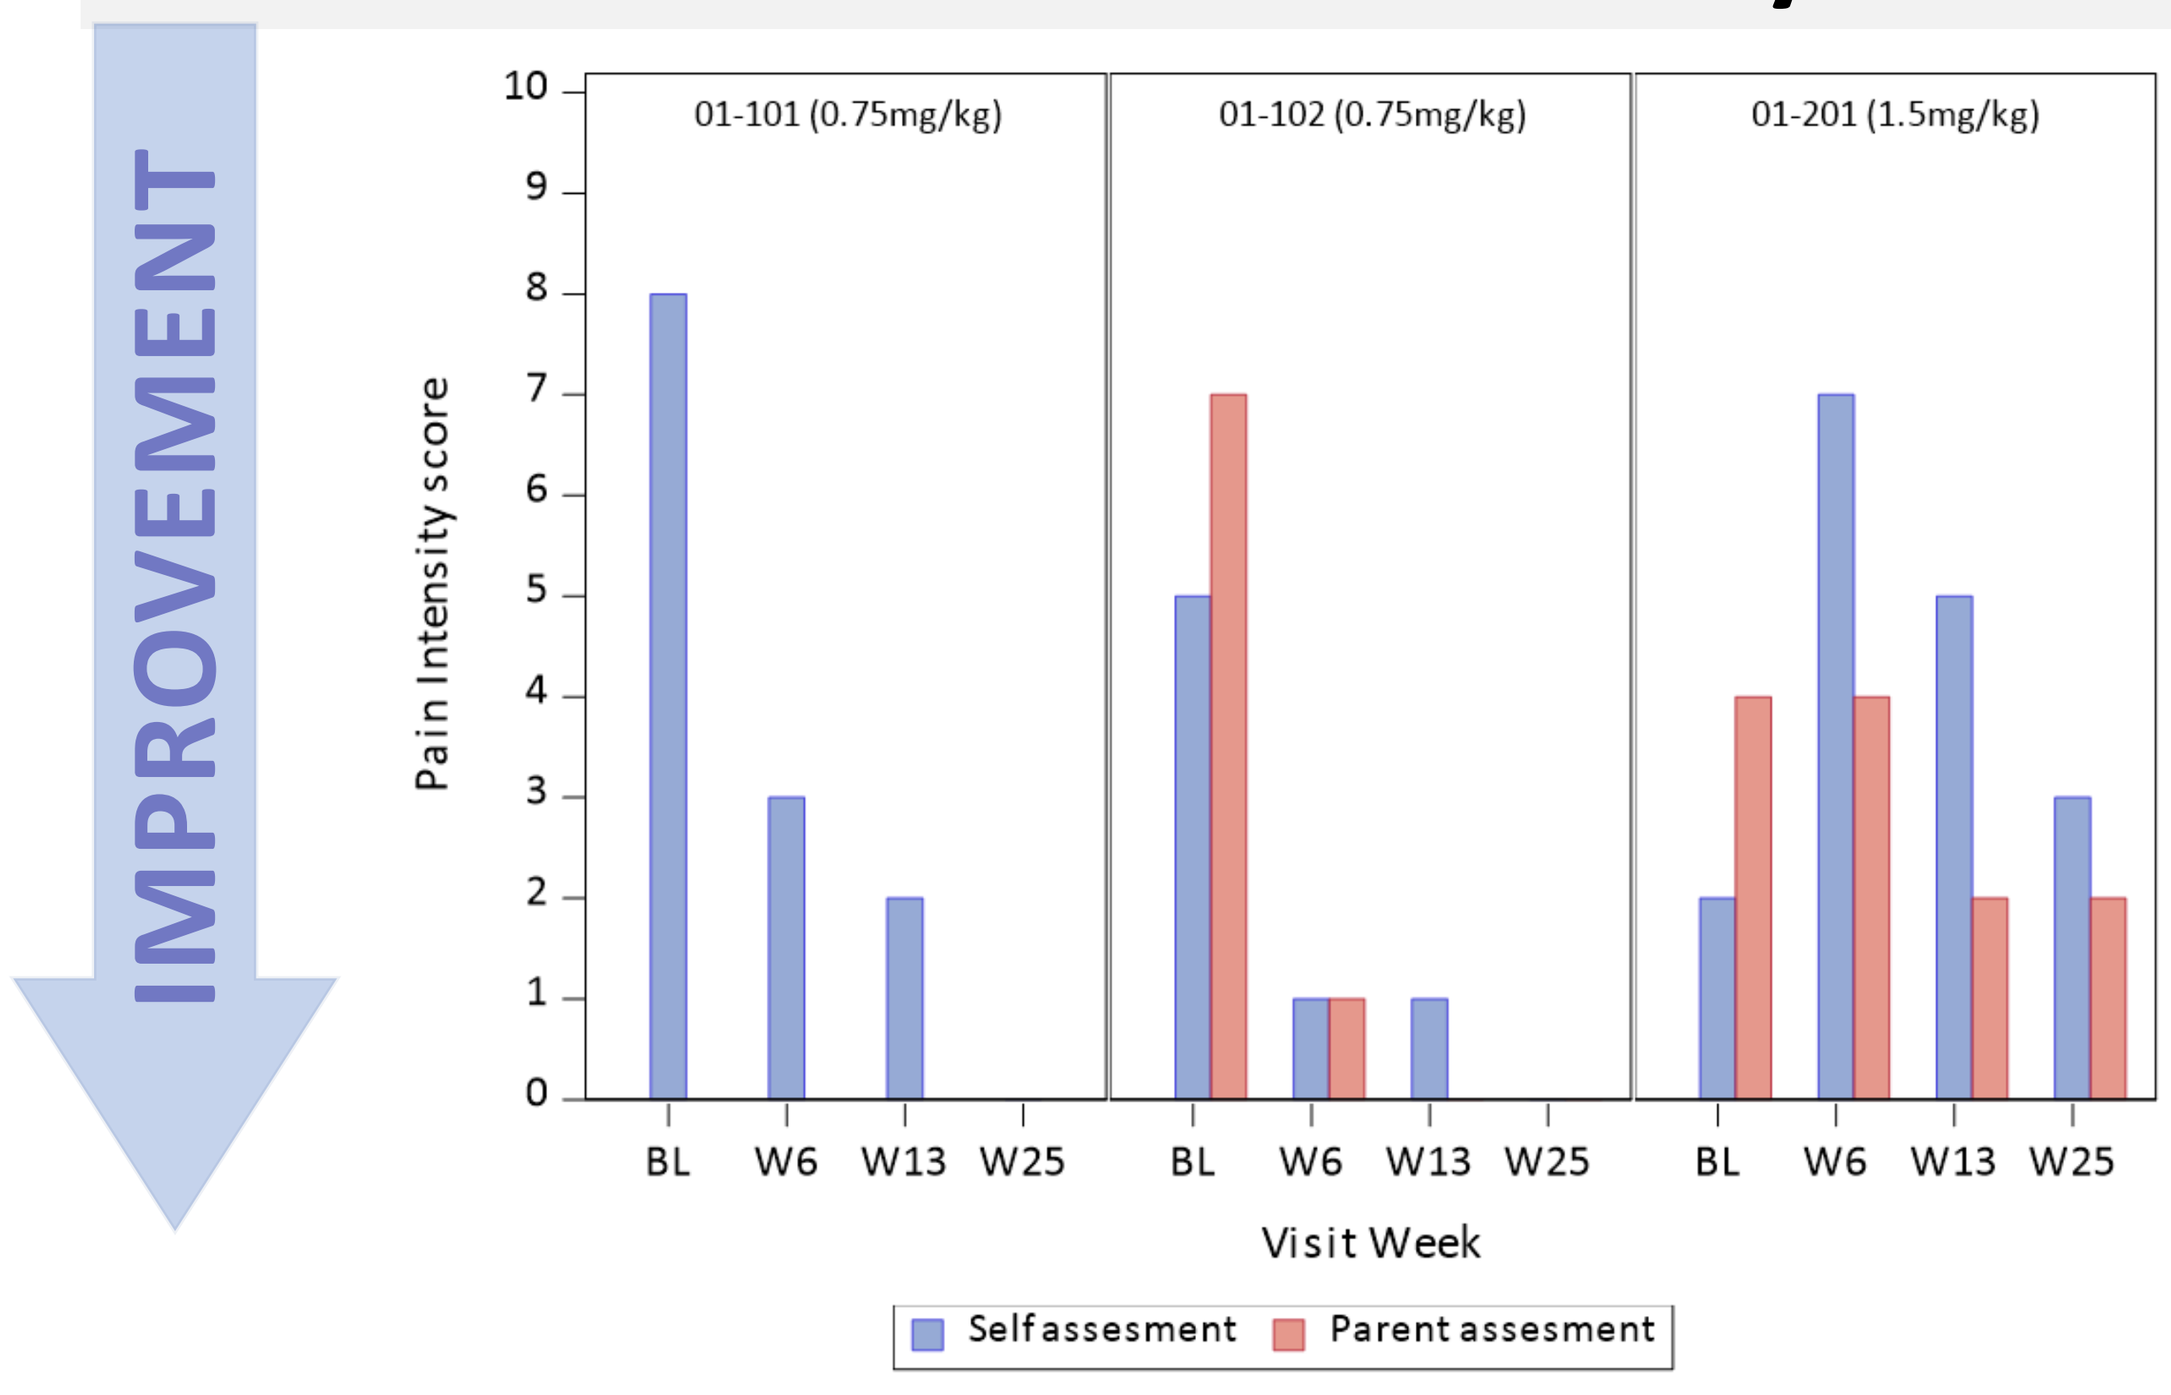

IMPROVEMENT
Pain Intensity score
01-101 (0.75mg/kg)
01-102 (0.75mg/kg)
01-201 (1.5mg/kg)
BL W6 W13 W25
Visit Week
Self assesment
Parent assesment

Subjects 01-101 and 01-102 felt no pain at week 25. 100% improvement noted for patients on 0.75mg/kg treatment, 50% improvement reported by parent proxy for patients on 1.5mg/kg

Pain Interference

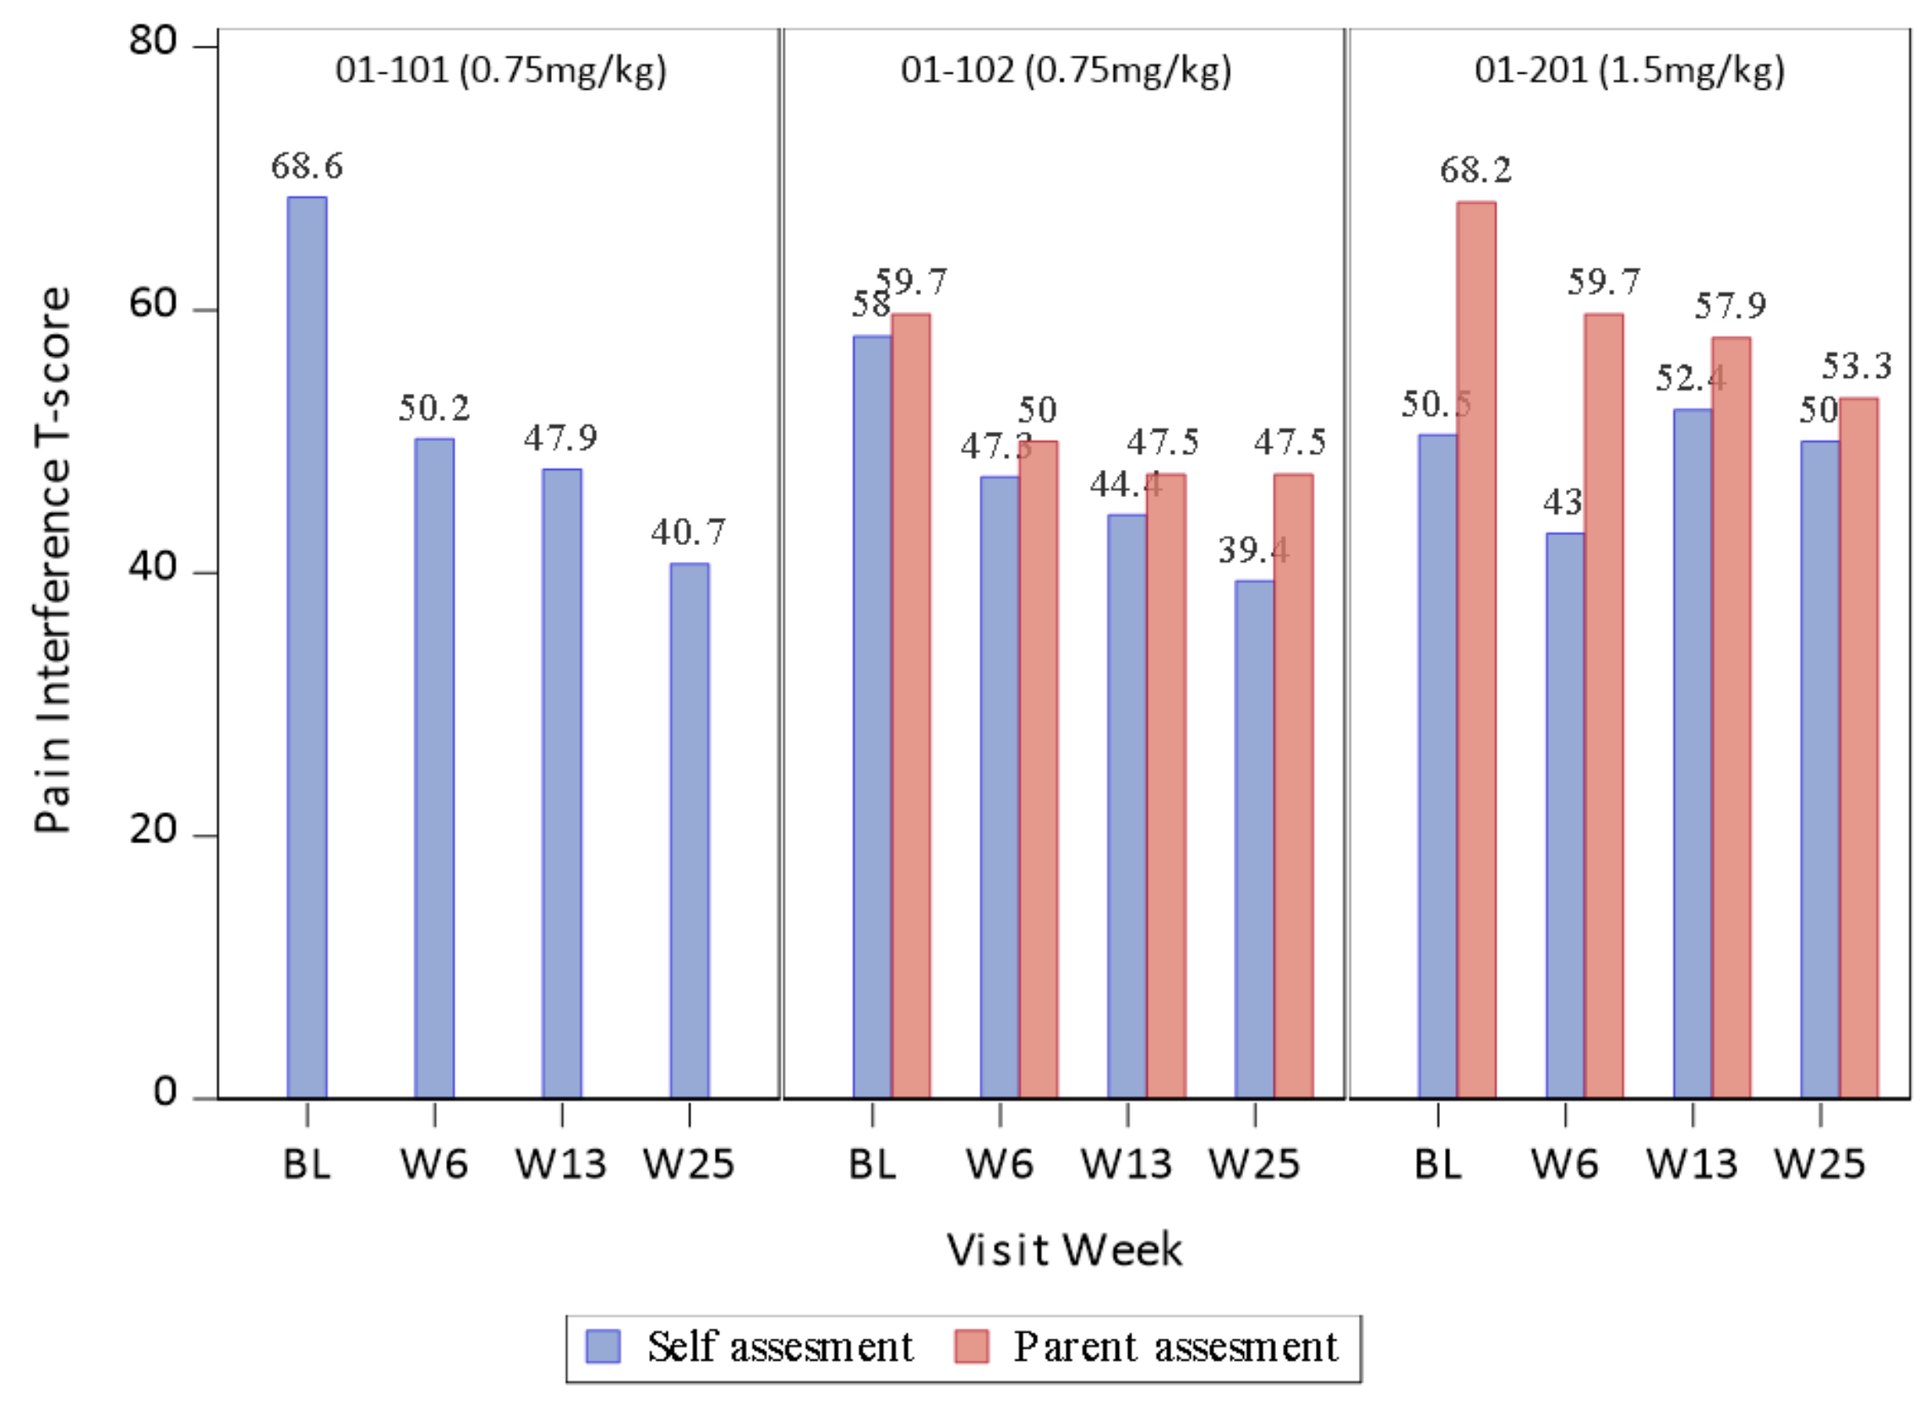

Pain Interference T-score
01-101 (0.75mg/kg)
01-102 (0.75mg/kg)
01-201 (1.5mg/kg)
68.6
50.2
47.9
40.7
58
59.7
47.3
50
44.4
47.5
39.4
47.5
50.5
68.2
43
59.7
52.4
57.9
50
53.3
BL W6 W13 W25
Visit Week
Self assesment
Parent assesment

Pain Interference: Symptom worst score is 80 and best score is 0, decrease indicates improvement.

- All patients or proxies demonstrated improvement in pain intensity and pain interference scores between baseline and week 25. Two patients had no pain at week 25.
- Exception was patient 01-201, whose baseline pain intensity assessment was significantly lower than subsequent assessments. This patient's pain intensity peaked at week 6 then gradually decreased.

PROMIS Pain Behaviour

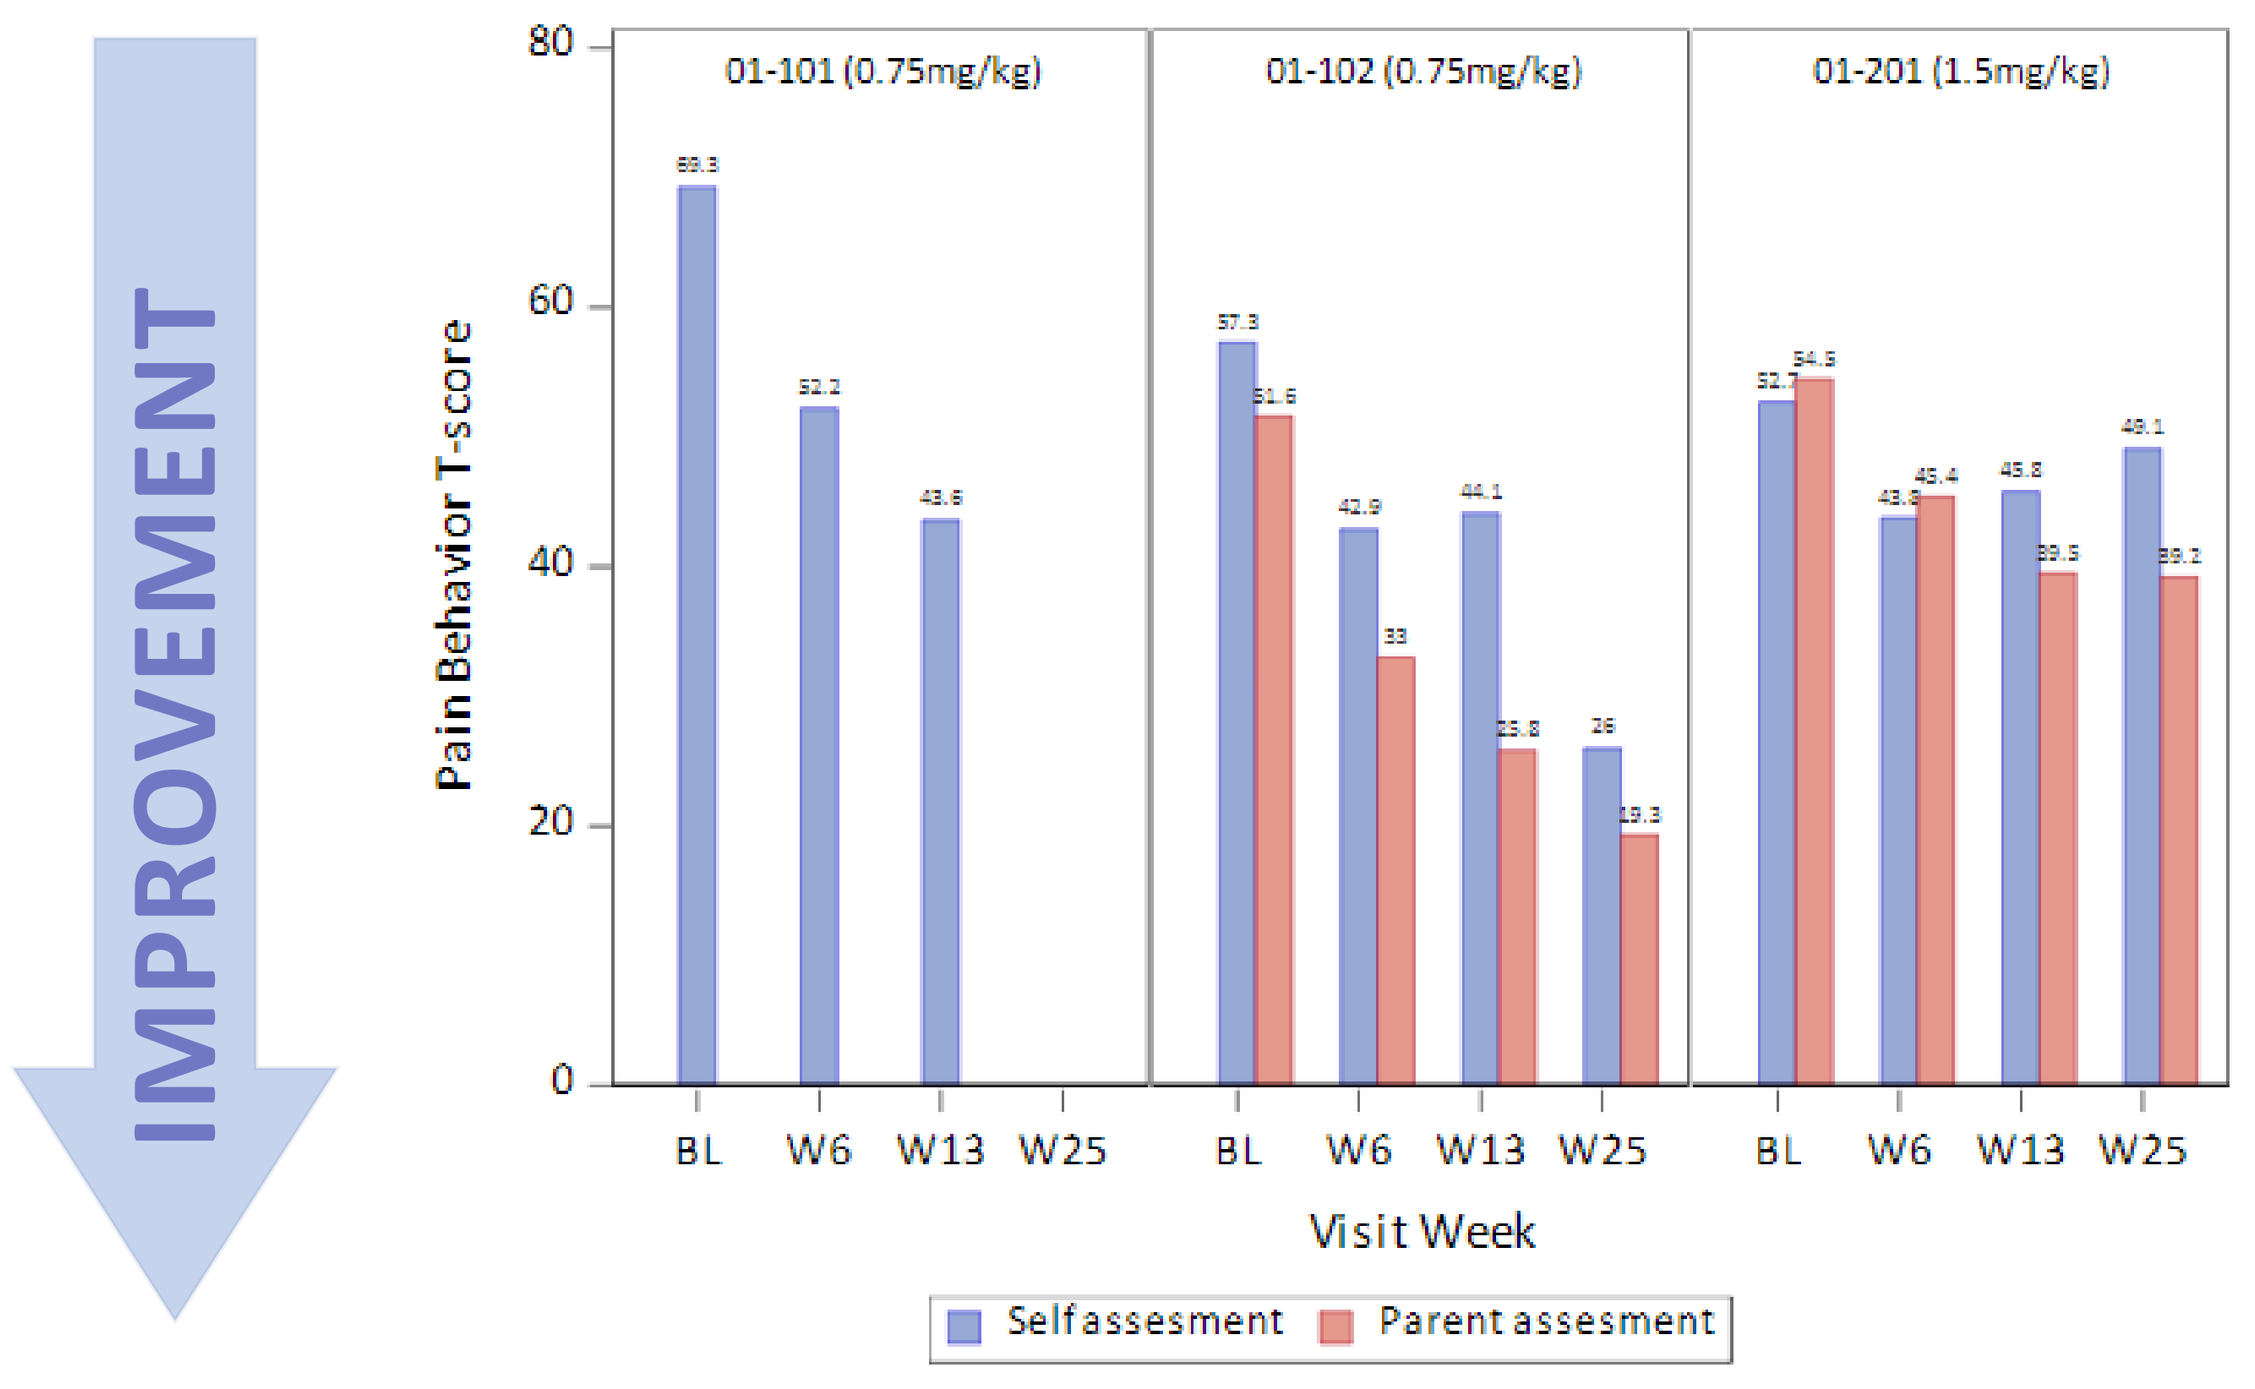

IMPROVEMENT
Pain Behavior T-score
01-101 (0.75mg/kg)
01-102 (0.75mg/kg)
01-201 (1.5mg/kg)
BL W6 W13 W25
Visit Week
Self assesment
Parent assesment

Pain Behavior: Symptom worst score is 80 and best score is 0, decrease indicates improvement. At week 25 subject 01-101 had no pain. Therefore, T-score was not calculated.

Fatigue

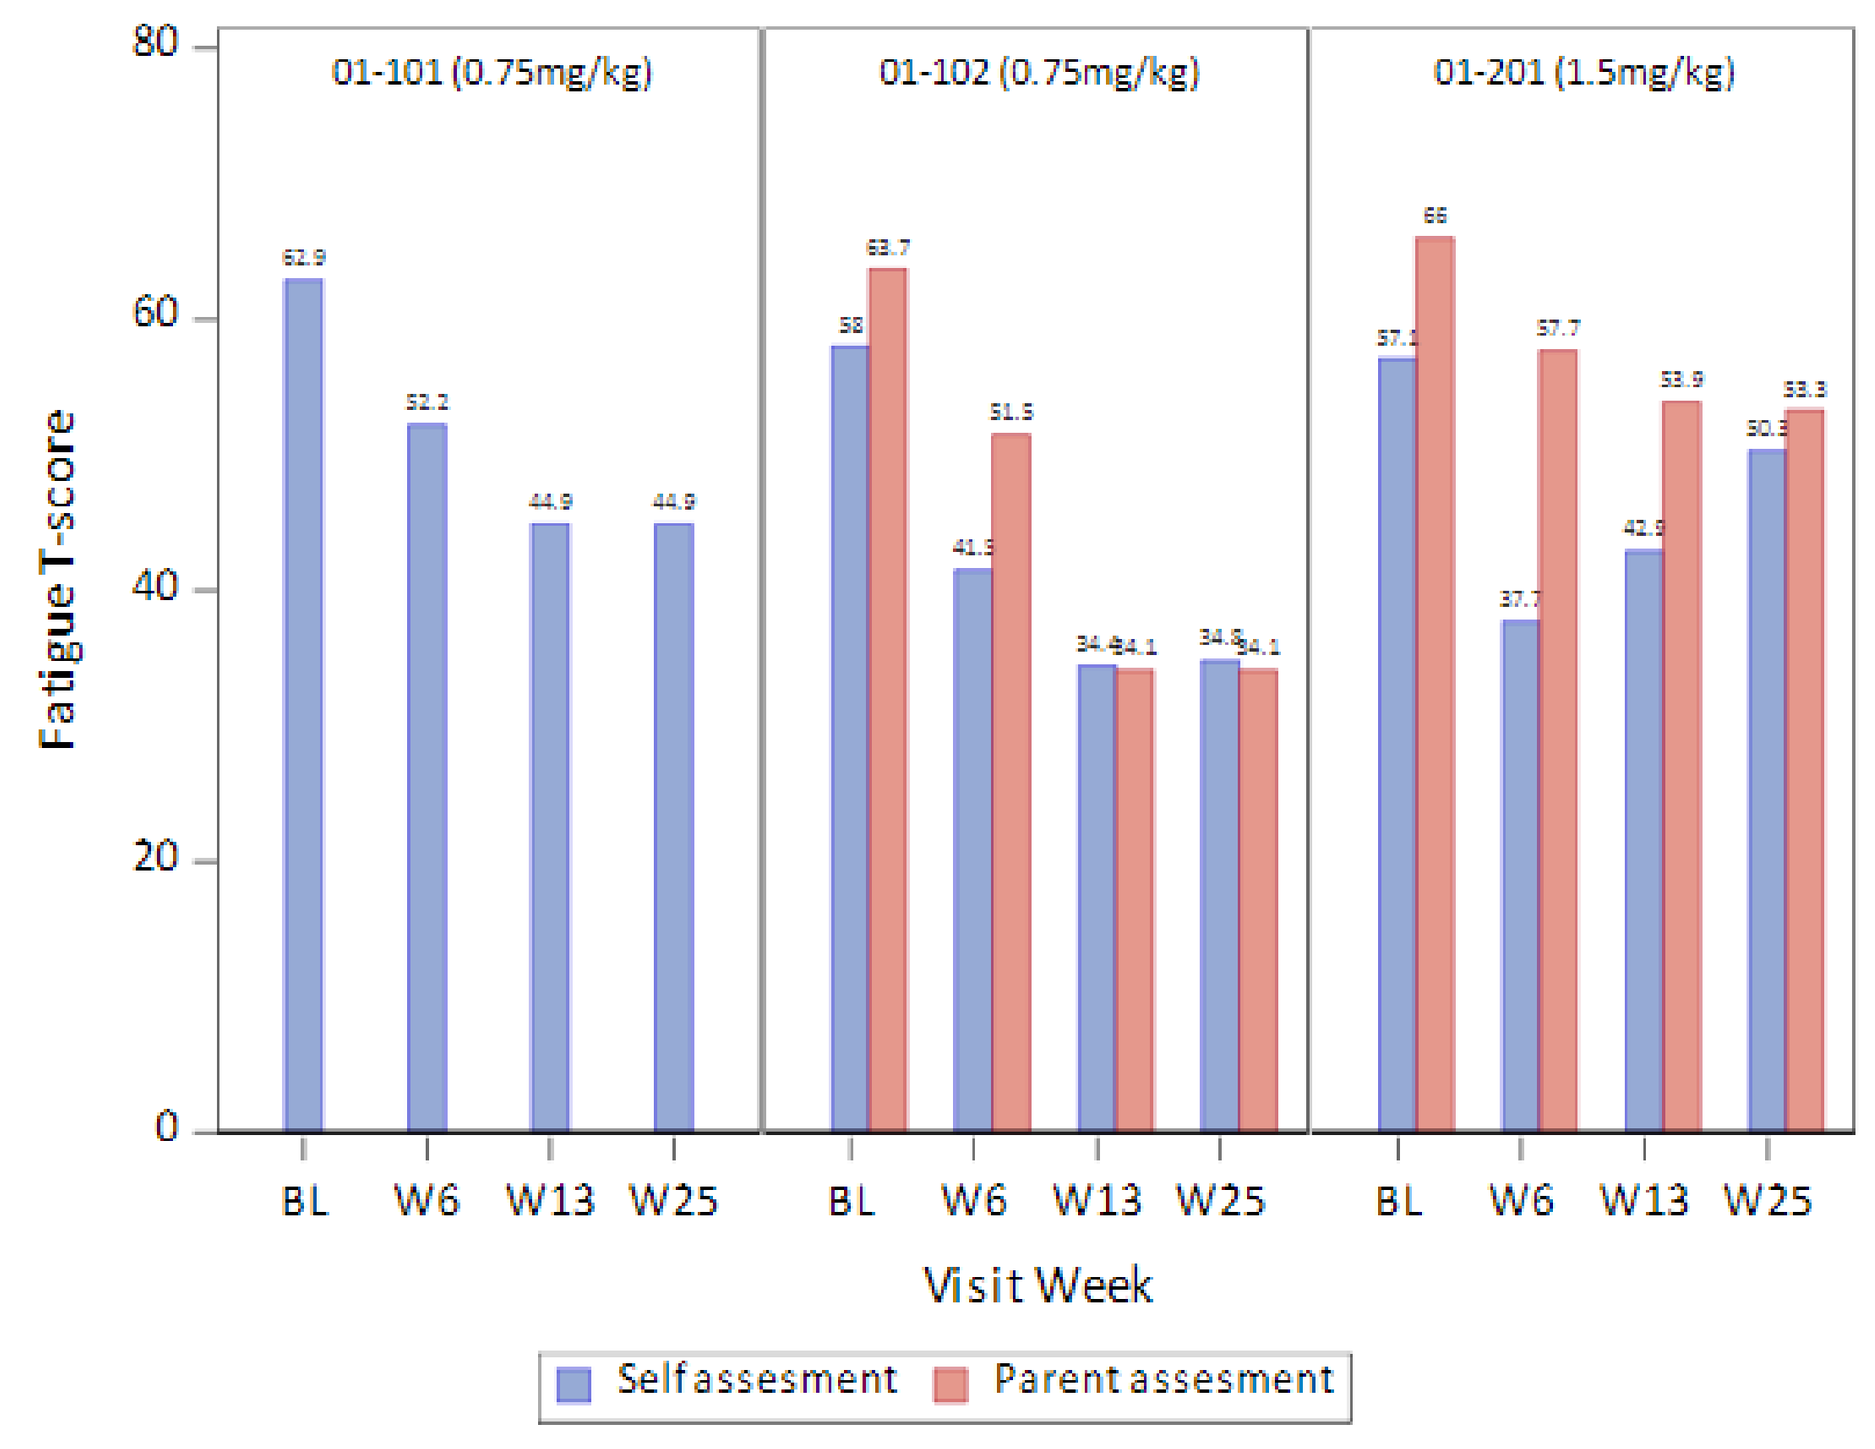

Fatigue T-score
01-101 (0.75mg/kg)
01-102 (0.75mg/kg)
01-201 (1.5mg/kg)
BL W6 W13 W25
Visit Week
Self assesment
Parent assesment

Fatigue: Symptom worst score is 80 and best score is 0, decrease indicates improvement.

- Both patients and proxies reported improvements in pain behaviour score out to week 25.
- At week 25, all patients experienced improvement in fatigue scores, which was mirrored by the proxy reports.

PROMIS Mobility

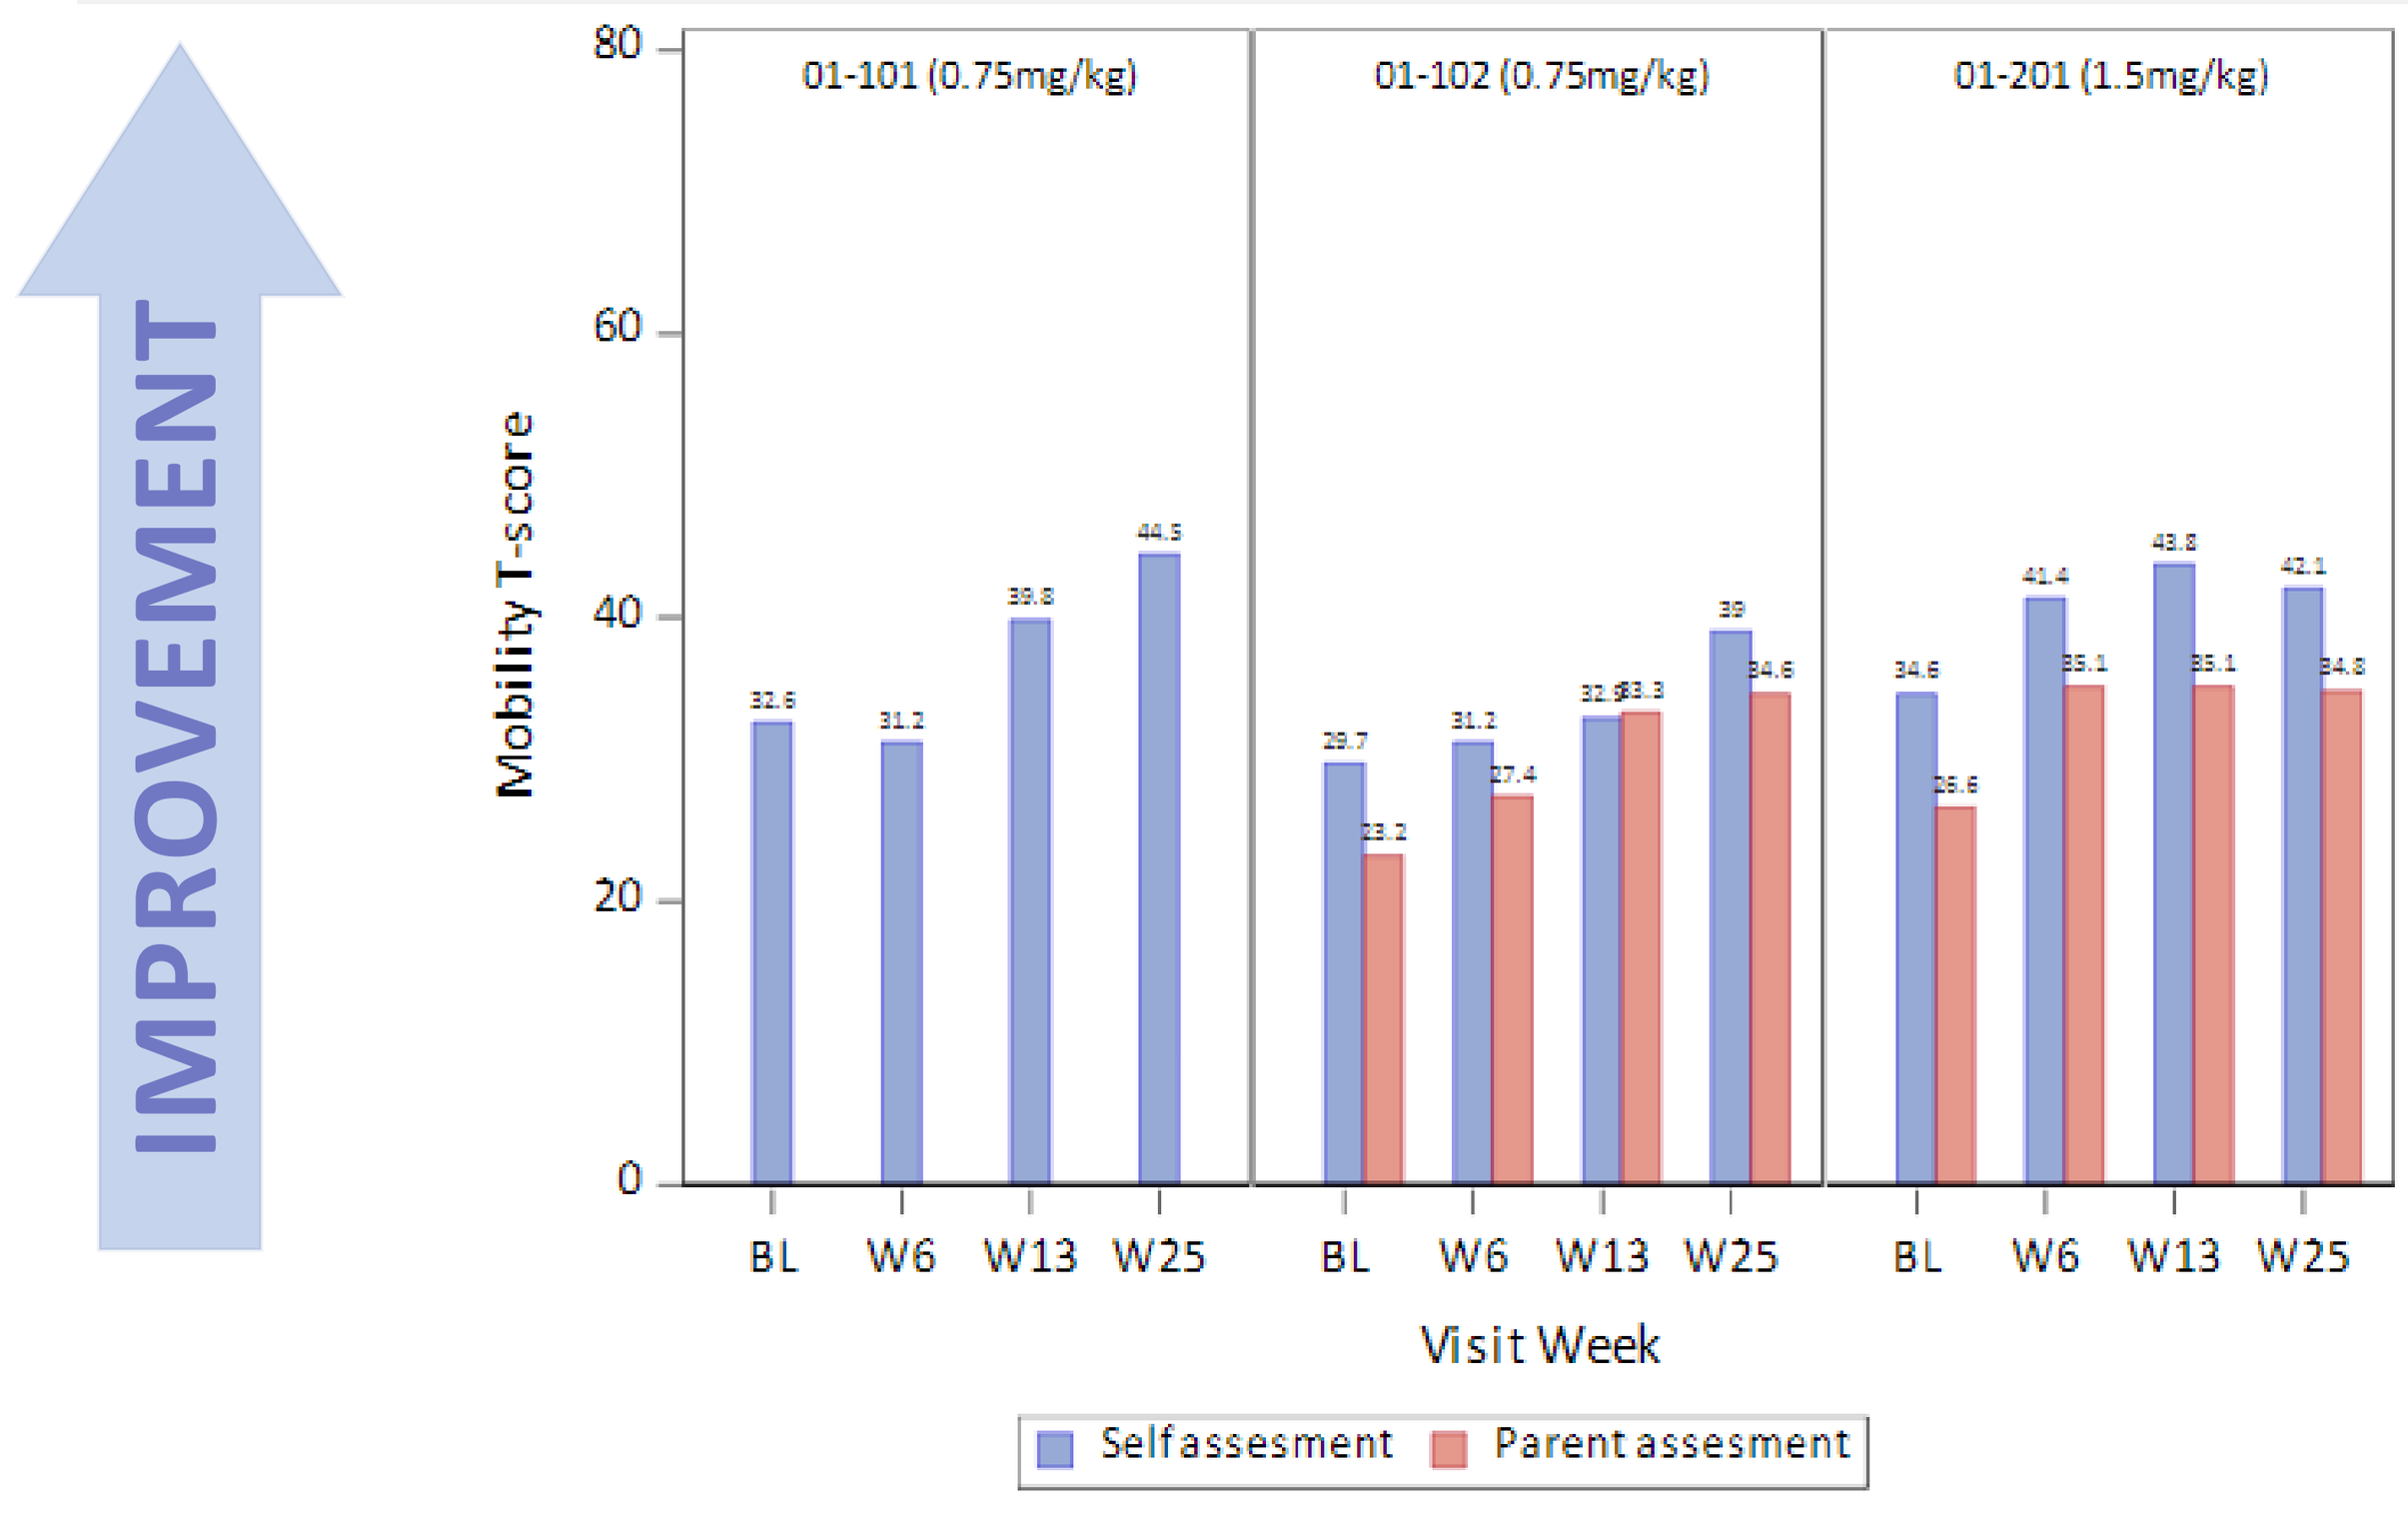

IMPROVEMENT
Mobility T-score
01-101 (0.75mg/kg)
01-102 (0.75mg/kg)
01-201 (1.5mg/kg)
BL W6 W13 W25
Visit Week
Self assesment
Parent assesment

Mobility: Function worst score is 0 and best score is 80, increase indicates improvement.

Upper Extremity

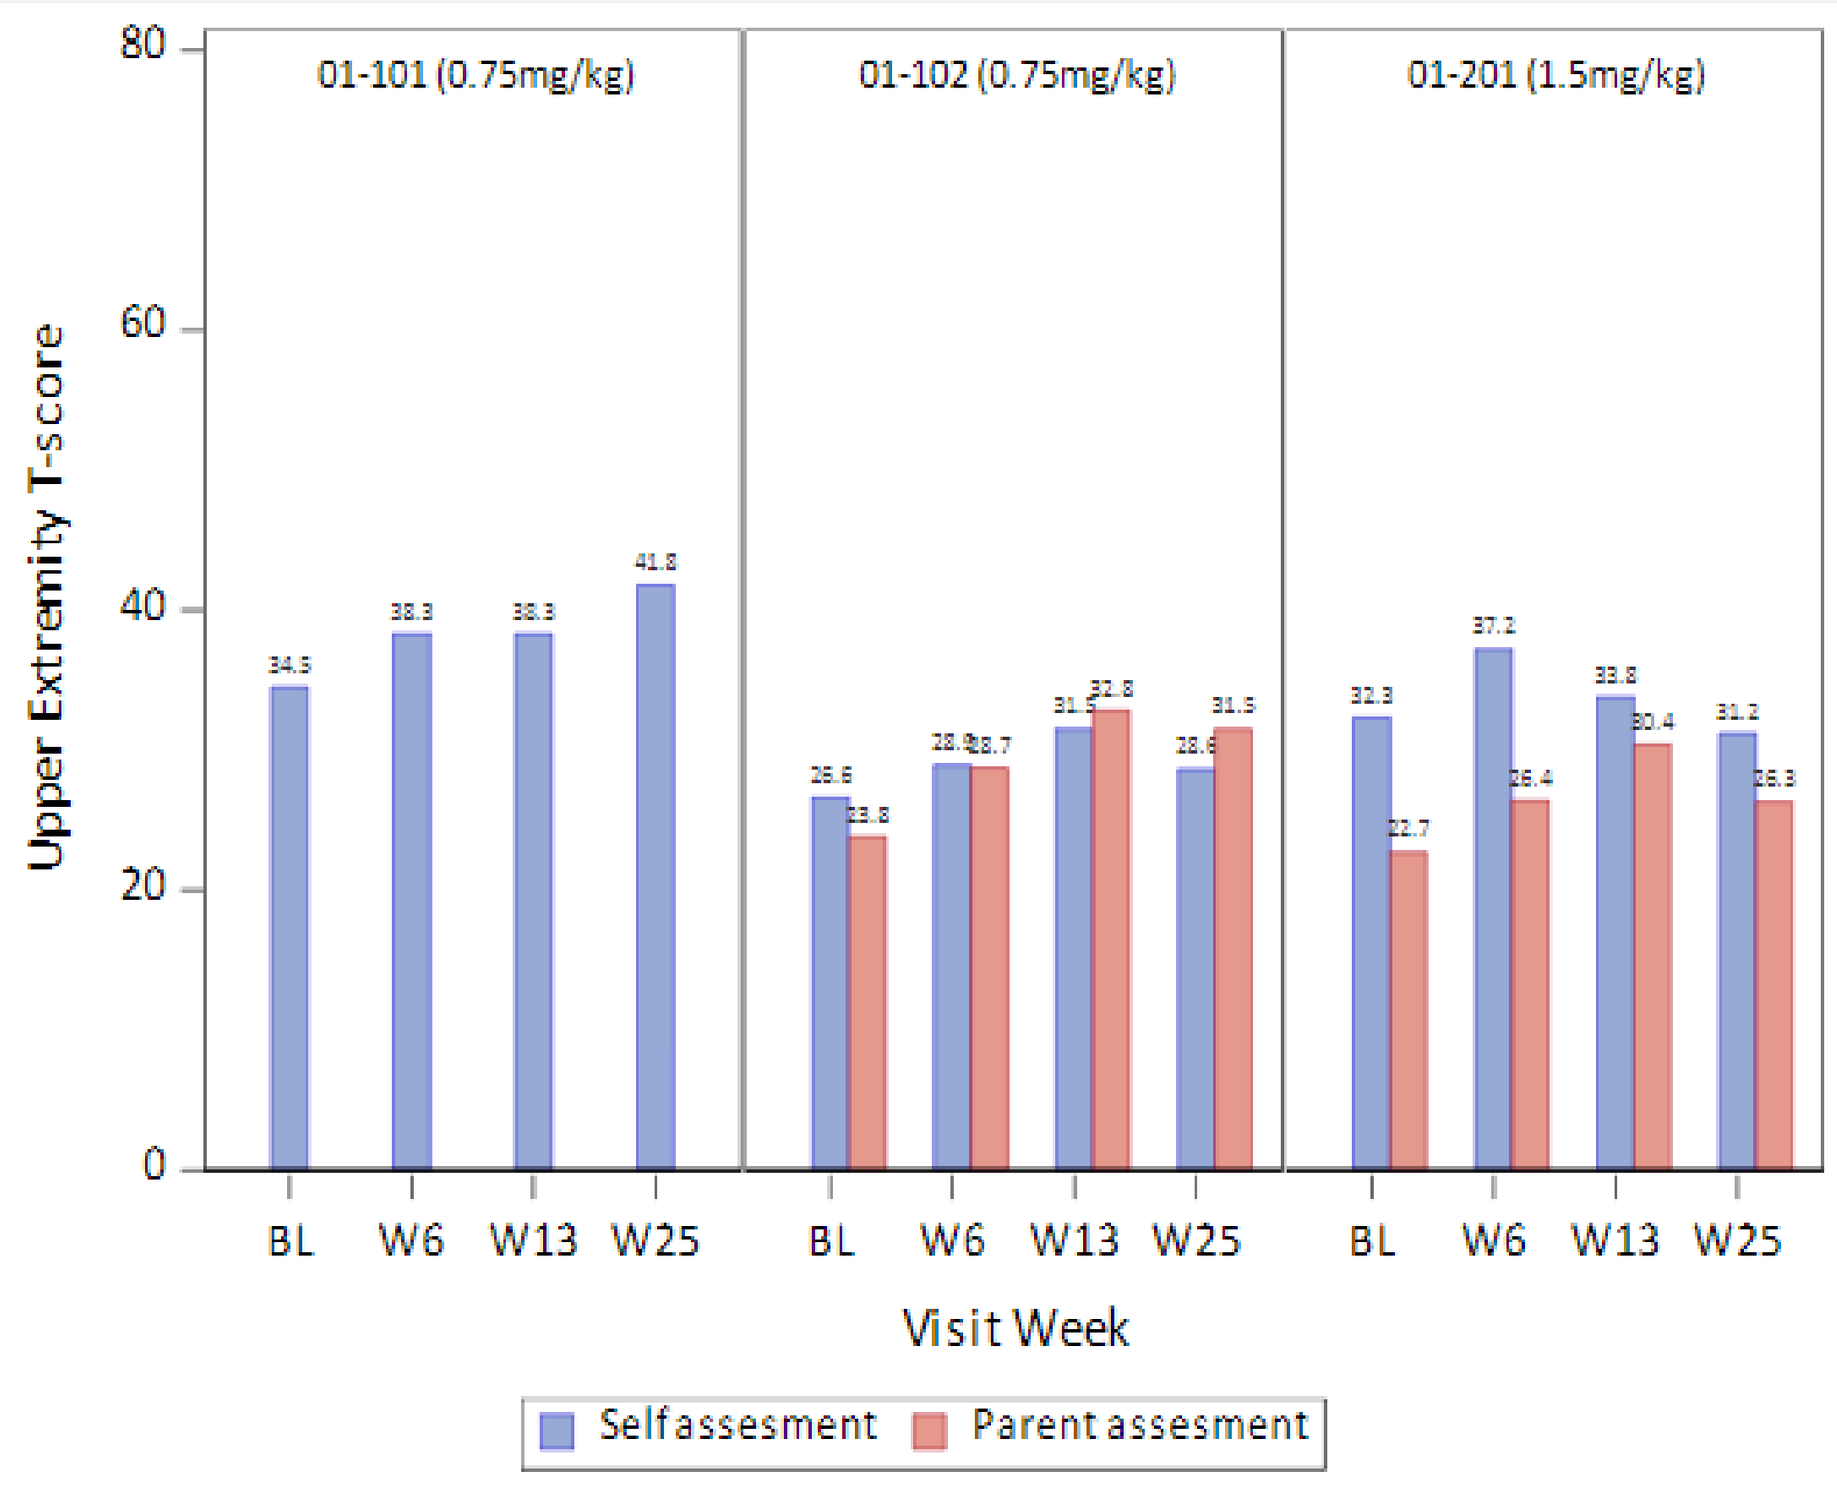

Upper Extremity T-score
01-101 (0.75mg/kg)
01-102 (0.75mg/kg)
01-201 (1.5mg/kg)
BL W6 W13 W25
Visit Week
Self assesment
Parent assesment

Upper Extremity: Function worst score is 0 and best score is 80, increase indicates improvement.

- Universally, all patients and proxies reported improved mobility scores by week 25.
- All proxies reported improvements in upper extremity (UE) function scores, although on self- report only 2 of the 3 patients reported improved UE function score at week 25.

PGIC

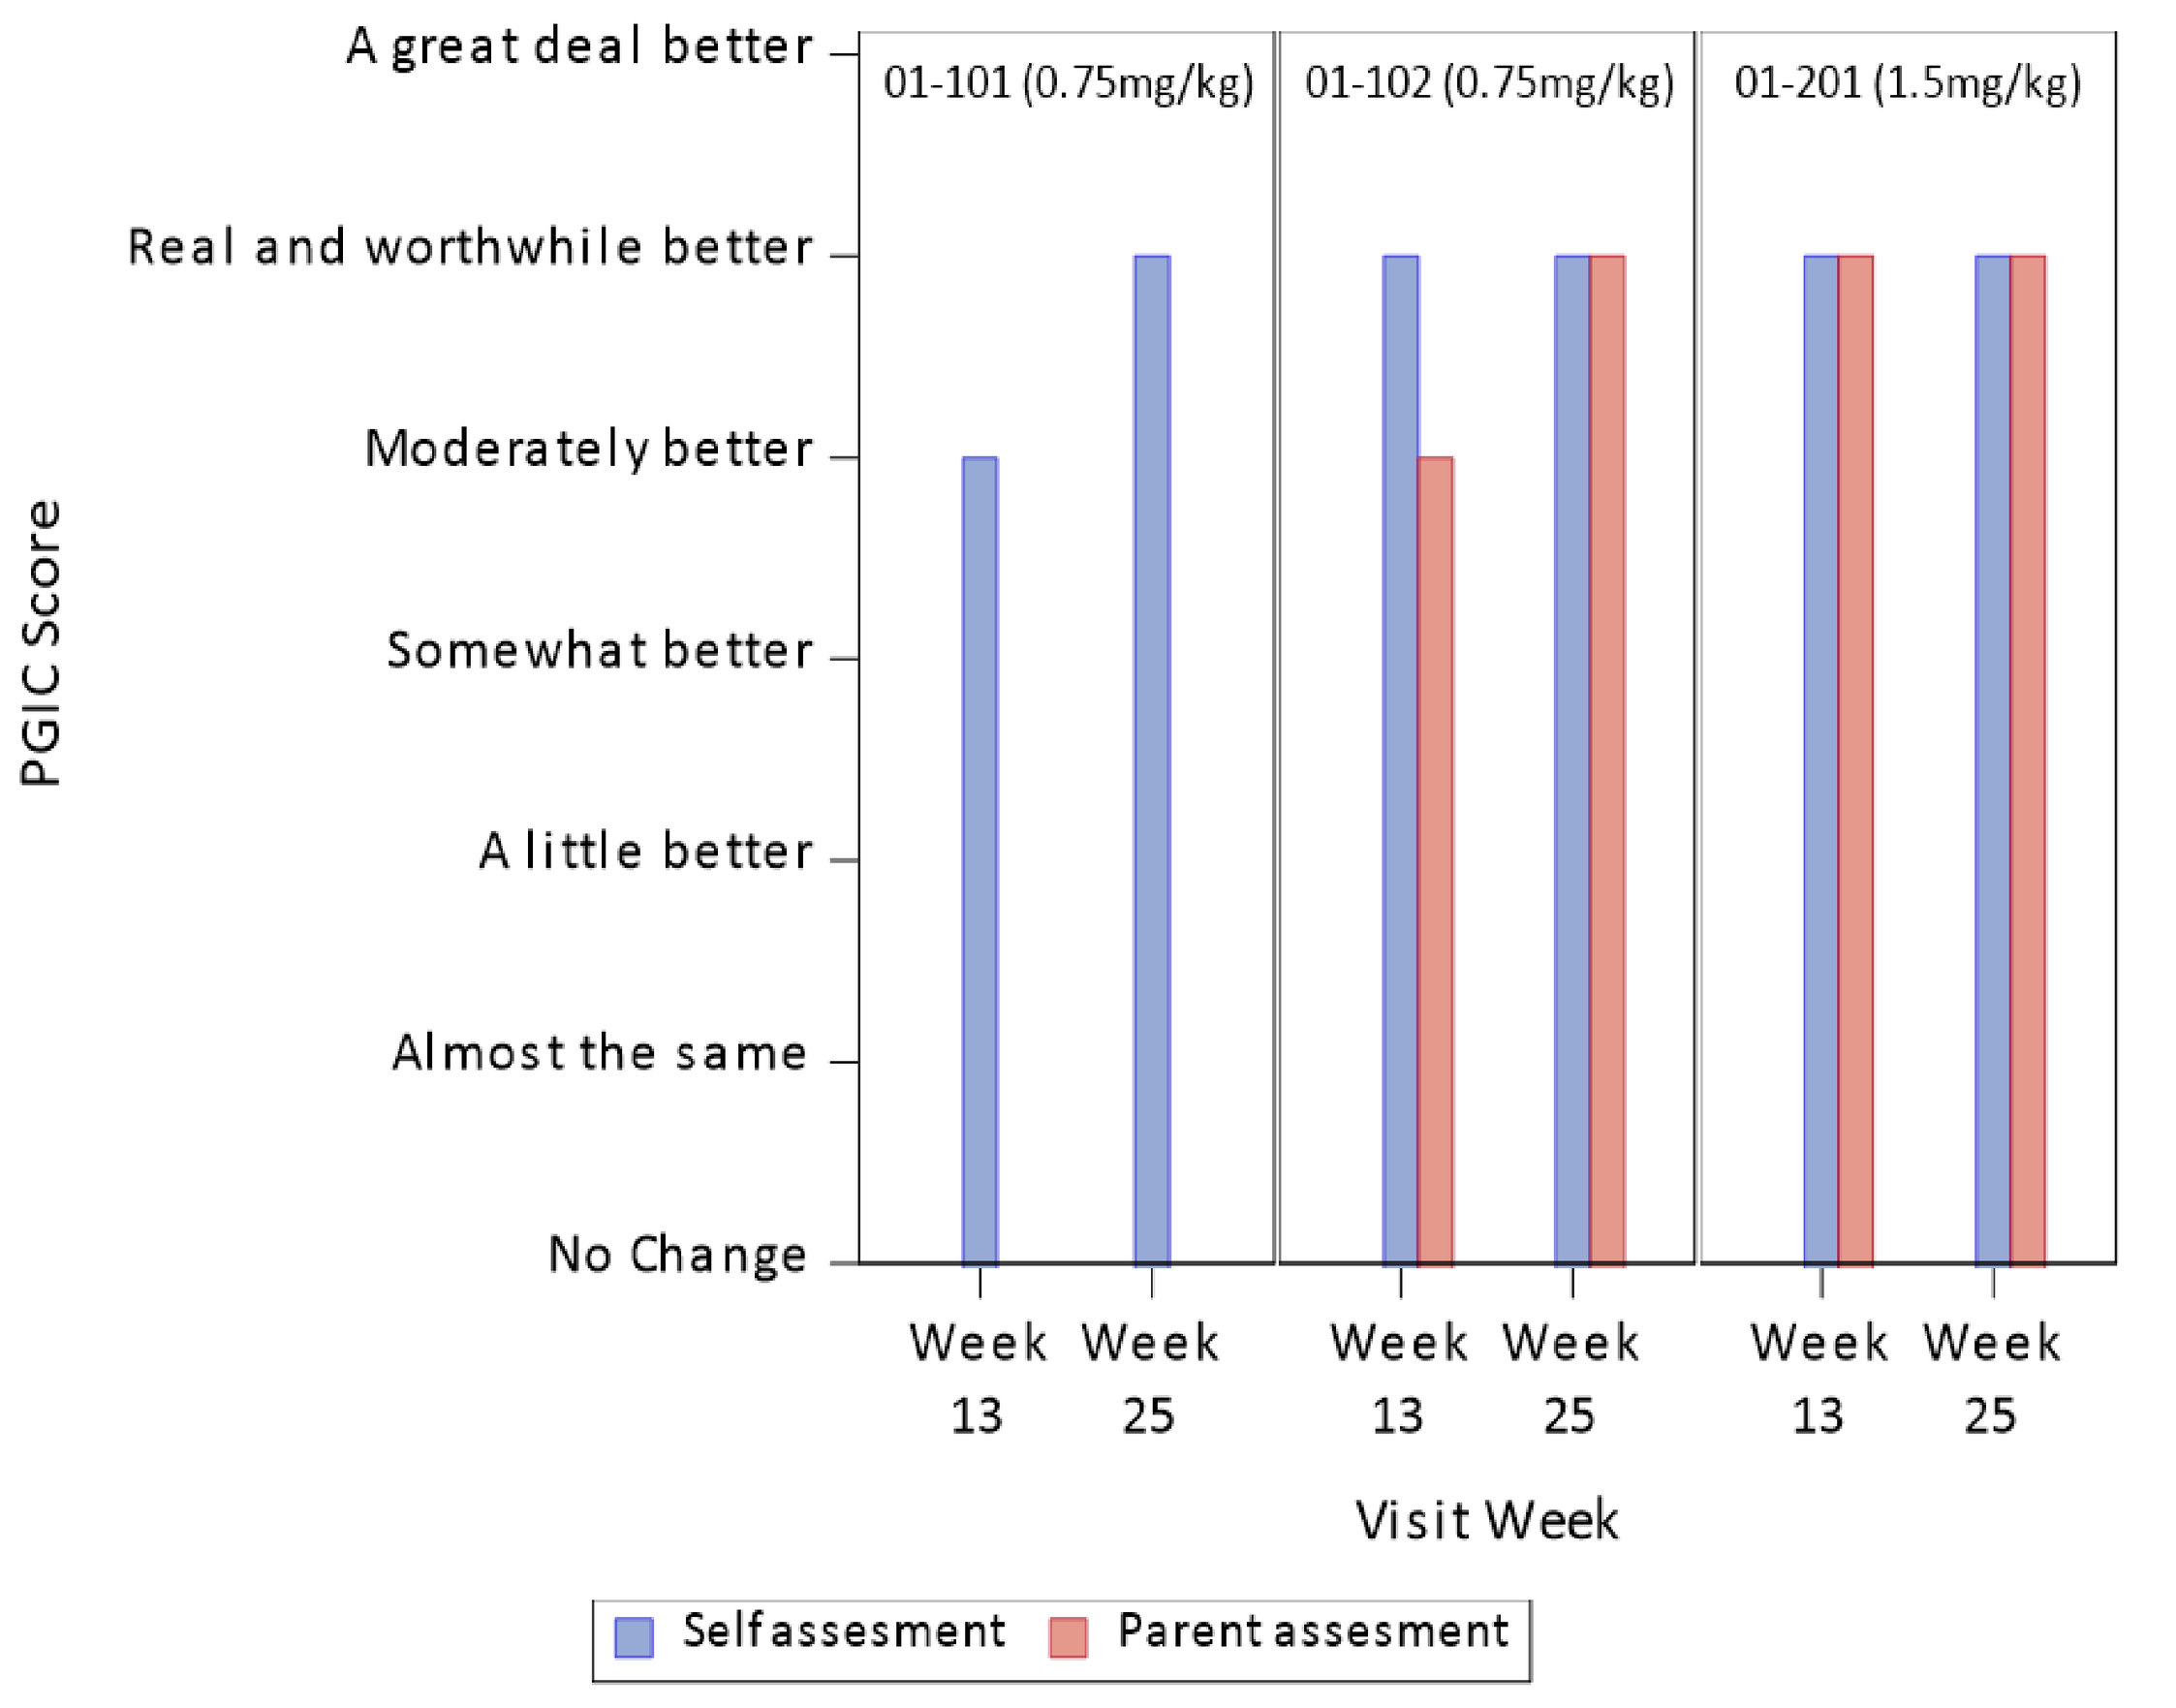

PGIC Score
A great deal better
Real and worthwhile better
Moderately better
Somewhat better
A little better
Almost the same
No Change
01-101 (0.75mg/kg)
01-102 (0.75mg/kg)
01-201 (1.5mg/kg)
Week 13
Week 25
Visit Week
Self assesment
Parent assesment

- At week 25, all patients and proxies reported improvement, reporting 'A definite improvement that has made a real and worthwhile difference'.

Conclusions

- Subcutaneous PPS was well tolerated at doses of 0.75 and 1.5 mg/kg out to 24 weeks of continuous dosing.
- There was an overall trend toward improvement in all domains assessed (with the exception of grip strength), providing for meaningful improvements in pain, function, and ADLs, and overall improvement in quality of life.
 - PPS administration resulted in improvements in 2MWT and 6MWT, Range of Motion, and GSGC, which includes a range of activities important to daily function in this population.
 - Clinically meaningful improvements in functional performance were supported by a range of PROs including pain intensity, interference and behaviour, and fatigue, as well as mobility and upper extremity function. This was supported by improvement in PGIC in all patients.
- PK assessments demonstrated consistency in serum concentrations that were dose-dependent. Urinary GAGs showed overall improvement in MPS I specific fragment.
- Changes in the profile of biomarkers suggest that PPS has the potential to modulate the inflammatory and joint degenerating biomarkers that are associated with arthralgia in MPS I patients.

MPS I patients continue to experience pain and arthropathy following HSCT and/or ERT. These often-debilitating symptoms significantly impact physical function and quality of life. PPS may address this unmet medical need and provide improvement in pain and function. Further studies are warranted to continue investigation of PPS in MPS I.

References

1 Valayannopoulos V, Nicely H, Harmatz P, et al. Mucopolysaccharidosis VI. Orphanet *J Rare Dis.* 2010;5:20.

2 Hendriksz CJ, Berger KI, Lampe C, et al. Health-related quality of life in mucopolysaccharidosis: looking beyond biomedical issues. Orphanet *J Rare Dis [Internet].* 2016;11:119. doi:10.1186/s13023-016-0503-2.

3 Harmatz P, Giugliani R, Schwartz I, et al. Enzyme replacement therapy for mucopolysaccharidosis VI: a phase 3, randomized, double-blind, placebo-controlled, multinational study of recombinant human N-acetylgalactosamine 4-sulfatase (recombinant human arylsulfatase B or rhASB) and follow-on, open-label extension study. *J Pediatr [Internet].* 2006;148:533–539. doi:10.1016/j.jpeds.2005.12.014.

Disclosures

PARA_MPSI_001 is funded by Paradigm Biopharmaceuticals Ltd. Michael Imperiale and Sharon Charles are employees of Paradigm Biopharmaceuticals Pty Ltd. The study was approved by WCHN Human Research Ethics Committee, and all patients provided written informed consent to participate in the study.

Funding

PARA_MPSI1_001 is funded by Paradigm Biopharmaceuticals Ltd. Paradigm Biopharmaceuticals Ltd is developing PPS for the treatment of complications of mucopolysaccharidosis including pain and function. The author(s) confirm(s) independence from the Sponsor; the content of this presentation has not been influenced by the Sponsor.

4 Hennermann JB, Gökce S, Solyom A, et al. Treatment with pentosan polysulphate in patients with MPS I: results from an open label, randomized, monocentric phase II study. *J Inherit Metab Dis* [Internet]. 2016;39:831–837. doi:10.1007/s10545-016-9974-5.

5 Orii K, Lim A, Tomatsu S, et al. Safety Study of Sodium Pentosan Polysulfate for Adult Patients with Mucopolysaccharidosis Type II. *Diagnostics* [Internet]. 2019;9:226. doi:10.3390/diagnostics9040226.

6 Furujo M, Orii K, Tomatsu S, et al. Safety evaluation of pentosan polysulfate for treatment of two Japanese siblings with mucopolysaccharidosis type VI in a phase 2 study. *Mol Genet Metab* [Internet]. 2019;126:S56–S57. doi:10.1016/j.ymgme.2018.12.130.

7 Dawn Phillips , Beth Leiro , Sharon Charles , Melanie Duiker, Paul Harmatz. Maroteaux-Lamy syndrome (Mucopolysaccharidosis type VI): symptoms and the impact on function and activities of daily living (ADL). Presented at 2020 WORLD Symposium.

8 Saville JT, McDermott BK, Fletcher JM, et al. Disease and subtype specific signatures enable precise diagnosis of the mucopolysaccharidoses. Genet Med [Internet]. 2019;21:753–757. doi:10.1038/s41436-018-0136-z.

Acknowledgements

The authors acknowledge and thank clinical trial patients and staff for their time and contribution to MPS I research. The authors acknowledge Maria Fuller, PhD, FFSc (RCPA), Clinical Scientist, Genetics and Molecular Pathology, SA Pathology, Adelaide; Michael A. Eldon, PhD, FCP, Partner, Expert Consultant in Clinical Pharmacology, Pharmacokinetics, and Pharmacometrics, NDA Partners, LLC; Beth Leiro, BScPT, Clinical Outcome Consultant; Catherine Stapledon, R&D and Translational Scientist at Paradigm; and Divya Navuru, Biometrics Manager at Paradigm.